Accelerating pancreatic cancer drug screening by leveraging genomics to select better *in vitro* models

Yoonjeong Cha[1], Joseph Perez-Rogers[2], Andrew Lysaght[1], Brian Weiner[1], Sarah Kolitz[1], Fadi Towfic[1], Kevin Fowler[1], Badri Vardarajan[3], Maxim Artyomov[4], Benjamin Zeskind [1], Rebecca Kusko[1]
1) Immuneering Corporation, Cambridge, MA.; 2) Boston University, Boston, MA; 3) Columbia University Medical Center, New York, NY; 4) Washington University, St. Louis, MO

Background

- The oncology clinical trial success rate is alarmingly low (13.4%)[1] despite urgent unmet clinical needs for new medicines.
- After many decades of major research and funding efforts, pancreatic cancer remains largely intractable with a dismal estimated 5-year survival rate of only 5-6%. [1]
- The projection that pancreatic cancer will be the second leading cause of cancer related death by 2030 [2] compounded by numerous clinical trial failures highlights a growing need for novel approaches to accelerate progress in new medicine development.
- Cell lines are frequently used for pre-clinical compound screening prior to animal models and human testing. Cell line selection criteria typically includes ease of access, robustness and literature prevalence rather than genomic background.
- Others have reported that the genomic derangements in the cell lines most commonly used for *in vitro* cancer studies may not be representative of what is found in cancer patient tumors [3].

Methods

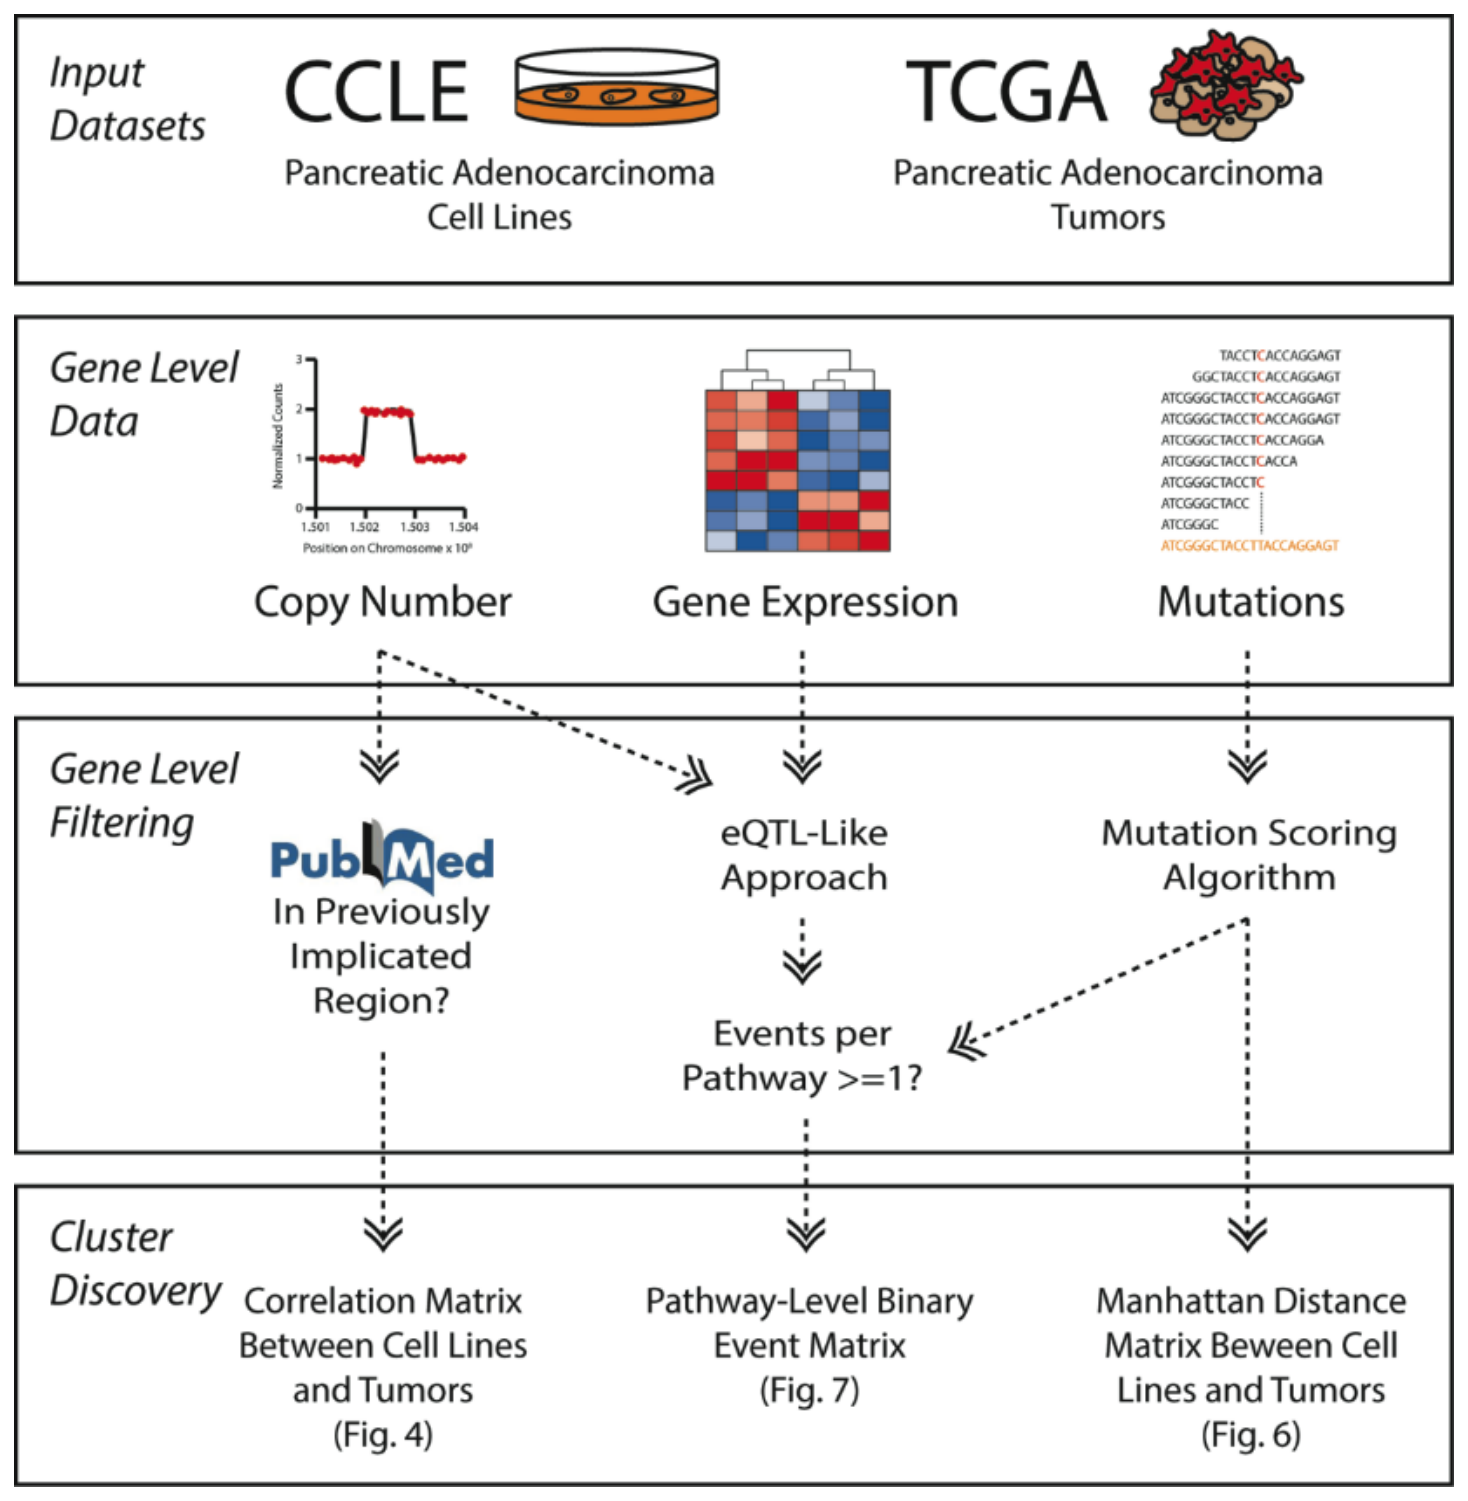
Input Datasets
CCLE
Pancreatic Adenocarcinoma Cell Lines
TCGA
Pancreatic Adenocarcinoma Tumors
Gene Level Data
Copy Number
Gene Expression
Mutations
Gene Level Filtering
PubMed
In Previously Implicated Region?
eQTL-Like Approach
Mutation Scoring Algorithm
Events per Pathway >=1?
Cluster Discovery
Correlation Matrix Between Cell Lines and Tumors (Fig. 4)
Pathway-Level Binary Event Matrix (Fig. 7)
Manhattan Distance Matrix Beween Cell Lines and Tumors (Fig. 6)

Figure 1. Methodology flowchart. In this study, we leveraged copy number variation (CNV), gene expression, and targeted mutation sequencing or exome data from 91 tumor samples from The Cancer Genome Atlas (TCGA) [4] and 44 cell lines from the Cancer Cell Line Encyclopedia (CCLE) [5] to predict optimal cell lines that mirror pancreatic cancer genomes most closely. CNVs with concurrent gene expression changes were compared to those found in cell lines. Mutations were filtered using various publicly available mutation scoring algorithms. Lastly, similarity between cell lines and mutations were scored using pathway analysis outlined below.

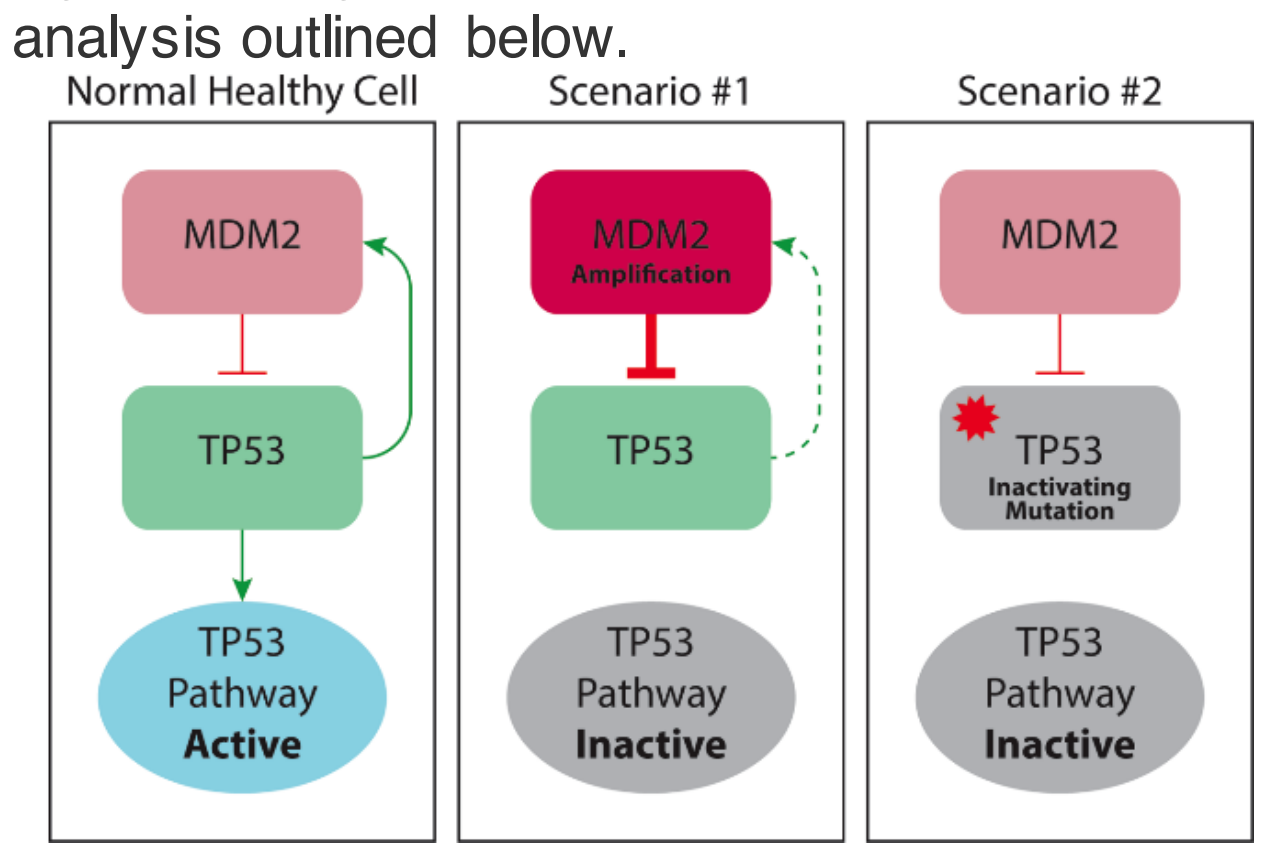
Normal Healthy Cell
Scenario #1
Scenario #2
MDM2
TP53
TP53 Pathway Active
MDM2 Amplification
TP53
TP53 Pathway Inactive
MDM2
TP53 Inactivating Mutation
TP53 Pathway Inactive

Figure 2. Rationale behind pathway summary methodology. Given that different alterations (CNVs or mutations) within the same pathway can have have the same end result, we chose to compare tumors to cell lines based on pathway perturbations in addition to directly comparing mutations and CNVs.

References

1. Siegel, Rebecca L., Kimberly D. Miller, and Ahmedin Jemal. "Cancer statistics, 2015." *CA: a cancer journal for clinicians* 65.1 (2015): 5-29.
2. Rahib, Lola, et al. "Projecting cancer incidence and deaths to 2030: the unexpected burden of thyroid, liver, and pancreas cancers in the United States." *Cancer research* 74.11 (2014): 2913-2921.
3. Domcke, Silvia, et al. "Evaluating cell lines as tumour models by comparison of genomic profiles." Nature communications 4 (2013).
4. http://cancergenome.nih.gov/
5. Barretina, Jordi, et al. "The Cancer Cell Line Encyclopedia enables predictive modelling of anticancer drug sensitivity." Nature 483.7391 (2012): 603-607.

Results

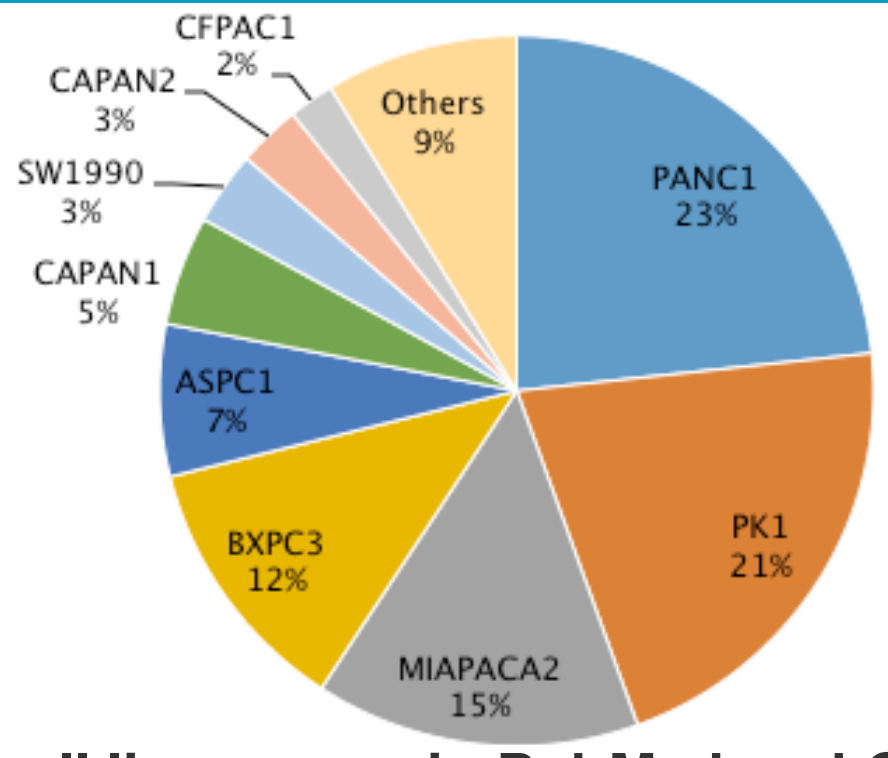
CFPAC1 2%
CAPAN2 3%
SW1990 3%
CAPAN1 5%
ASPC1 7%
BXPC3 12%
MIAPACA2 15%
PK1 21%
PANC1 23%
Others 9%

Figure 3. Searching cell line names in PubMed and Google Scholar for all cell lines in CCLE shows a strong bias towards certain cell lines compared to others. PANC1, PK1, MIAPACA2, BXPC3, ASPC1, CAPAN1, SW1990, CAPAN2, CFPAC1 account for 91% of the total citations. The remaining 9% includes 35 cell lines: SUIT2, T3M4, HPAC, HPAFII, PSN1, HS766T, QCP1, DANG, KLM1, PL45, KP4, L33, PATU8902, KP3, PATU8988T, YAPC, KP2, PATU8988S, PK45H, PK59, HUPT3, HUPT4, SU8686, PANC0327, SNU213, SNU410, KCIMOH1, PANC0403, TCCPAN2, PANC1005, SNU324, PANC0203, PANC0213, PANC0504, and PANC0813. Pi chart represents percent of total hits.

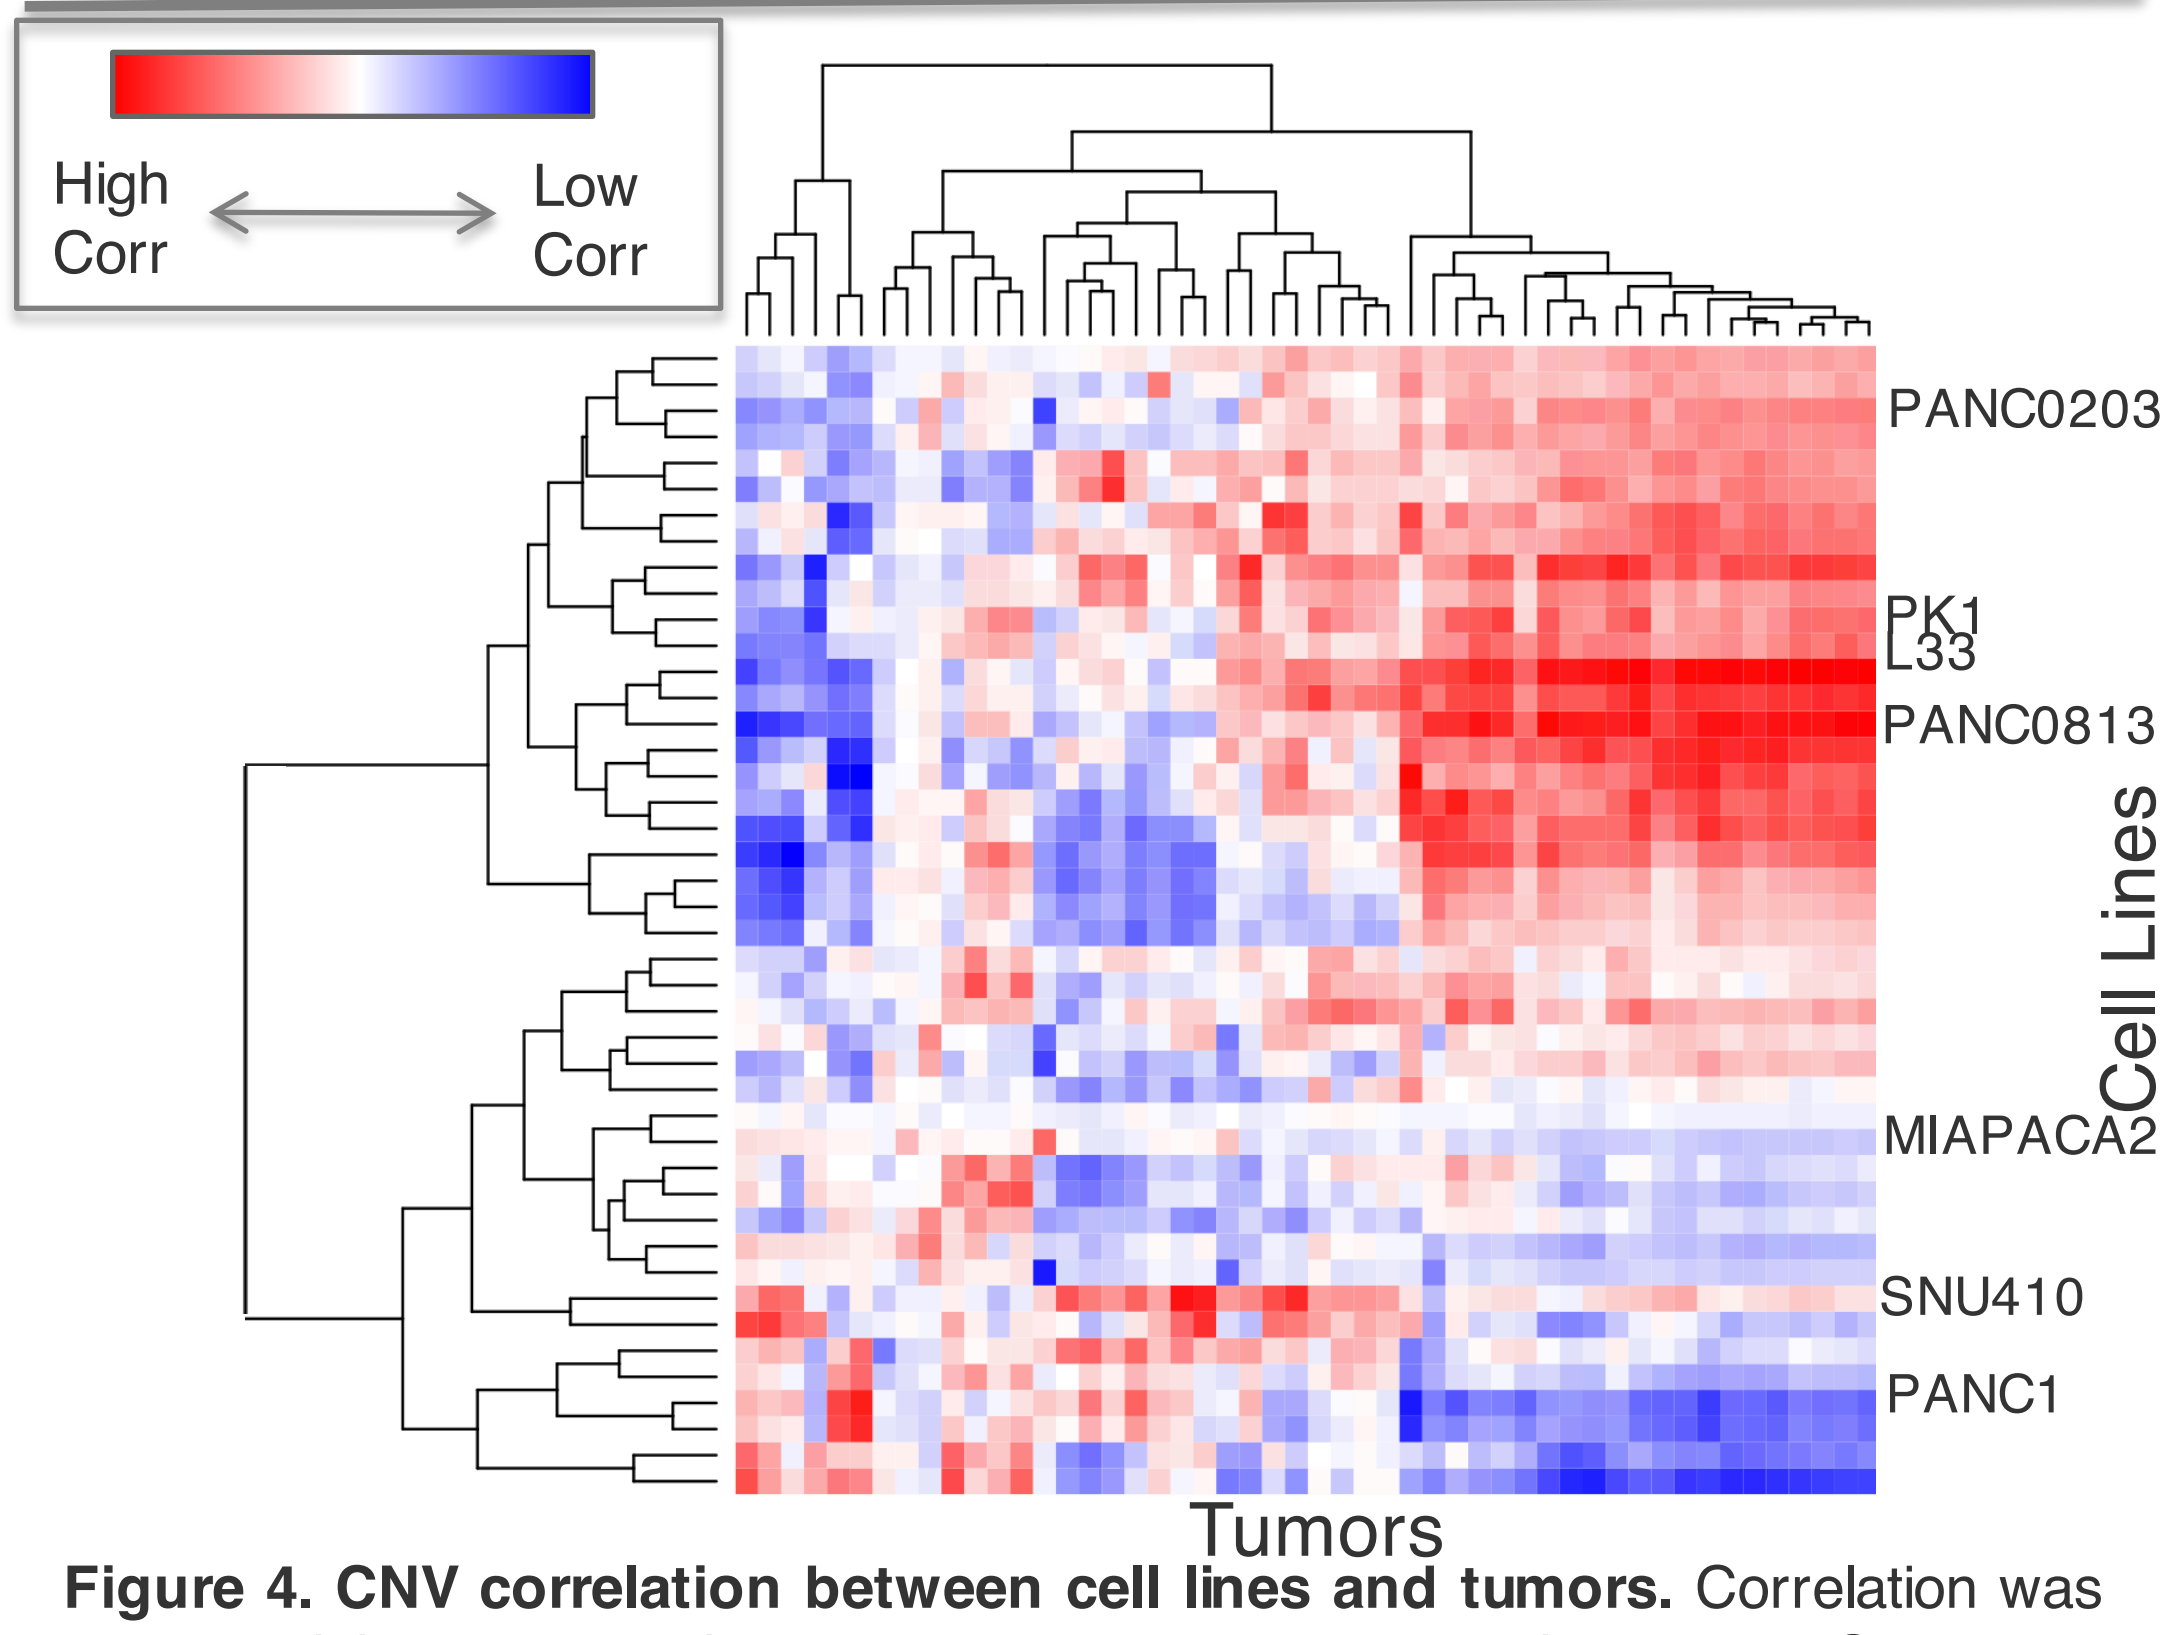
High Corr
Low Corr
PANC0203
PK1
L33
PANC0813
MIAPACA2
SNU410
PANC1
Cell Lines
Tumors

Figure 4. CNV correlation between cell lines and tumors. Correlation was calculated between each pancreatic cancer tumor and cell line. Some cell lines appear to have good correlation with many tumors whereas others do not. Overall, it was observed that cell lines have more CNVs than tumors, representing an important limitation of cell lines as models for tumors.

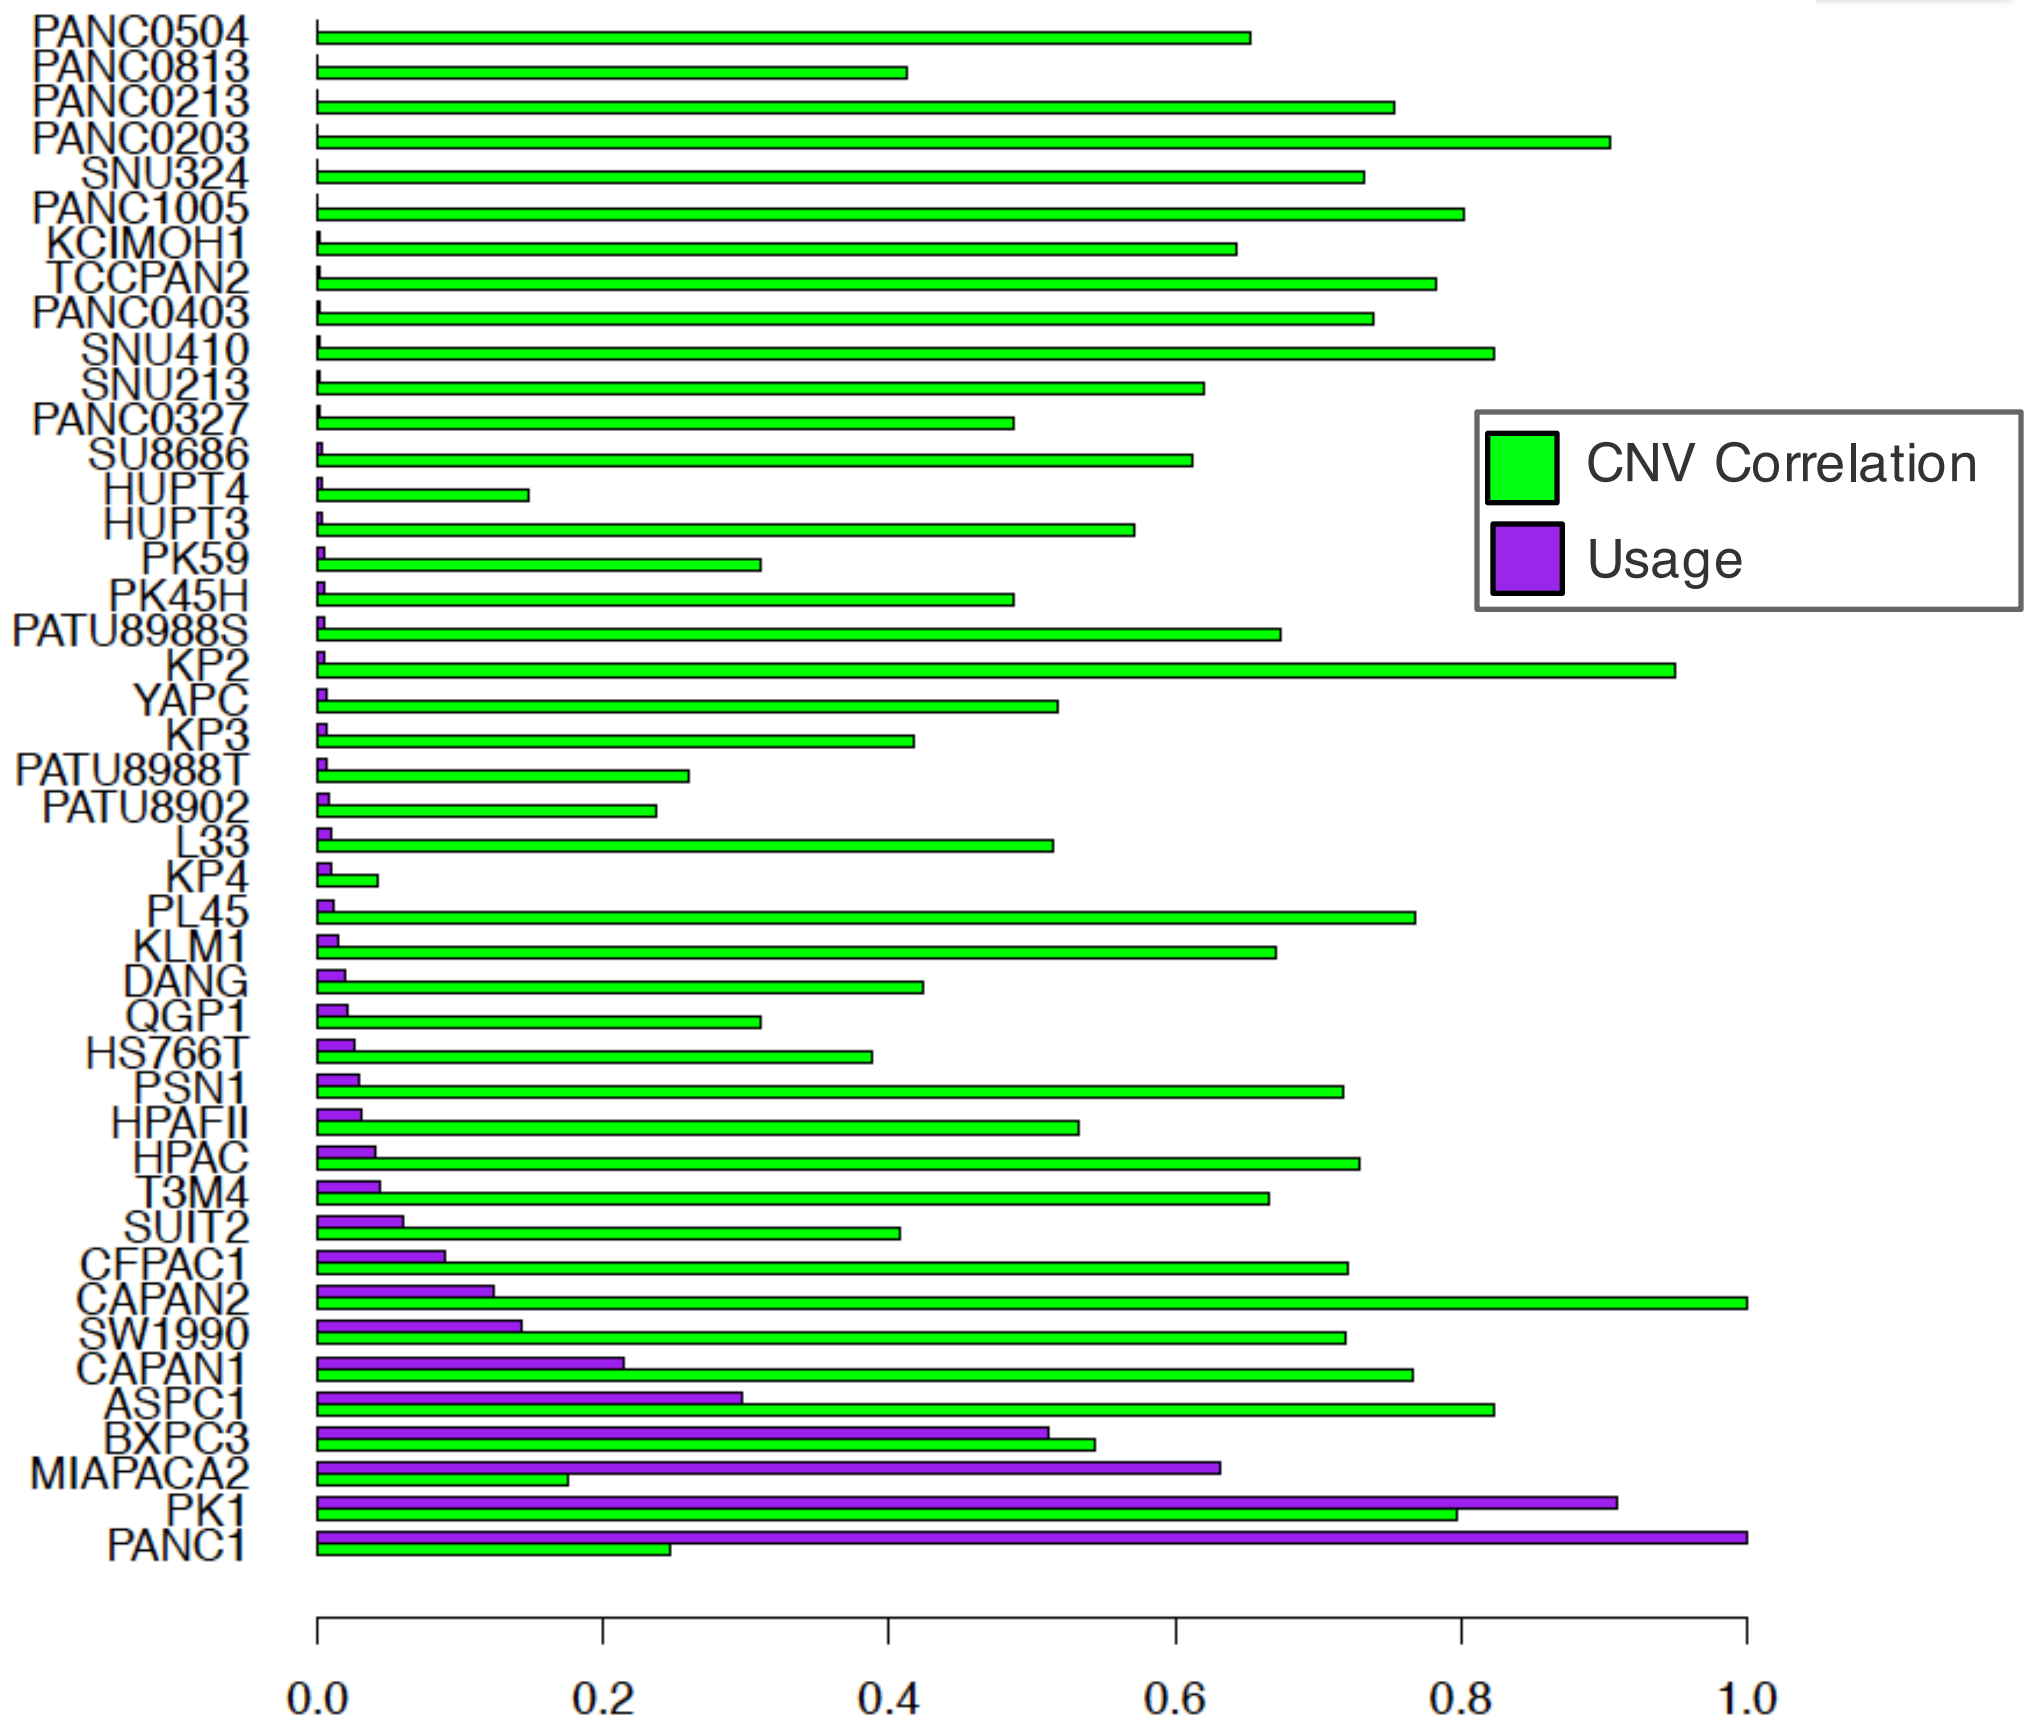
PANC0504
PANC0813
PANC0213
PANC0203
SNU324
PANC1005
KCIMOH1
TCCPAN2
PANC0403
SNU410
SNU213
PANC0327
SU8686
HUPT4
HUPT3
PK59
PK45H
PATU8988S
KP2
YAPC
KP3
PATU8988T
PATU8902
L33
KP4
PL45
KLM1
DANG
QGP1
HS766T
PSN1
HPAFII
HPAC
T3M4
SUIT2
CFPAC1
CAPAN2
SW1990
CAPAN1
ASPC1
BXPC3
MIAPACA2
PK1
PANC1
CNV Correlation
Usage
0.0
0.2
0.4
0.6
0.8
1.0

Figure 5. Comparing the median per gene CNV values in TCGA pancreatic cancer tumors and pancreatic cancer cell lines in CCLE. Alarmingly, the top five cell lines by CNV correlation with TCGA pancreatic tumors represented less than 10% out of all literature search hits for all pancreatic cancer cell lines, indicating that the most commonly used cell lines are not optimal from a genomics perspective. To bring CNV correlation and literature popularity to the same scale, both were divided by their respective max value.

Results (cont'd)

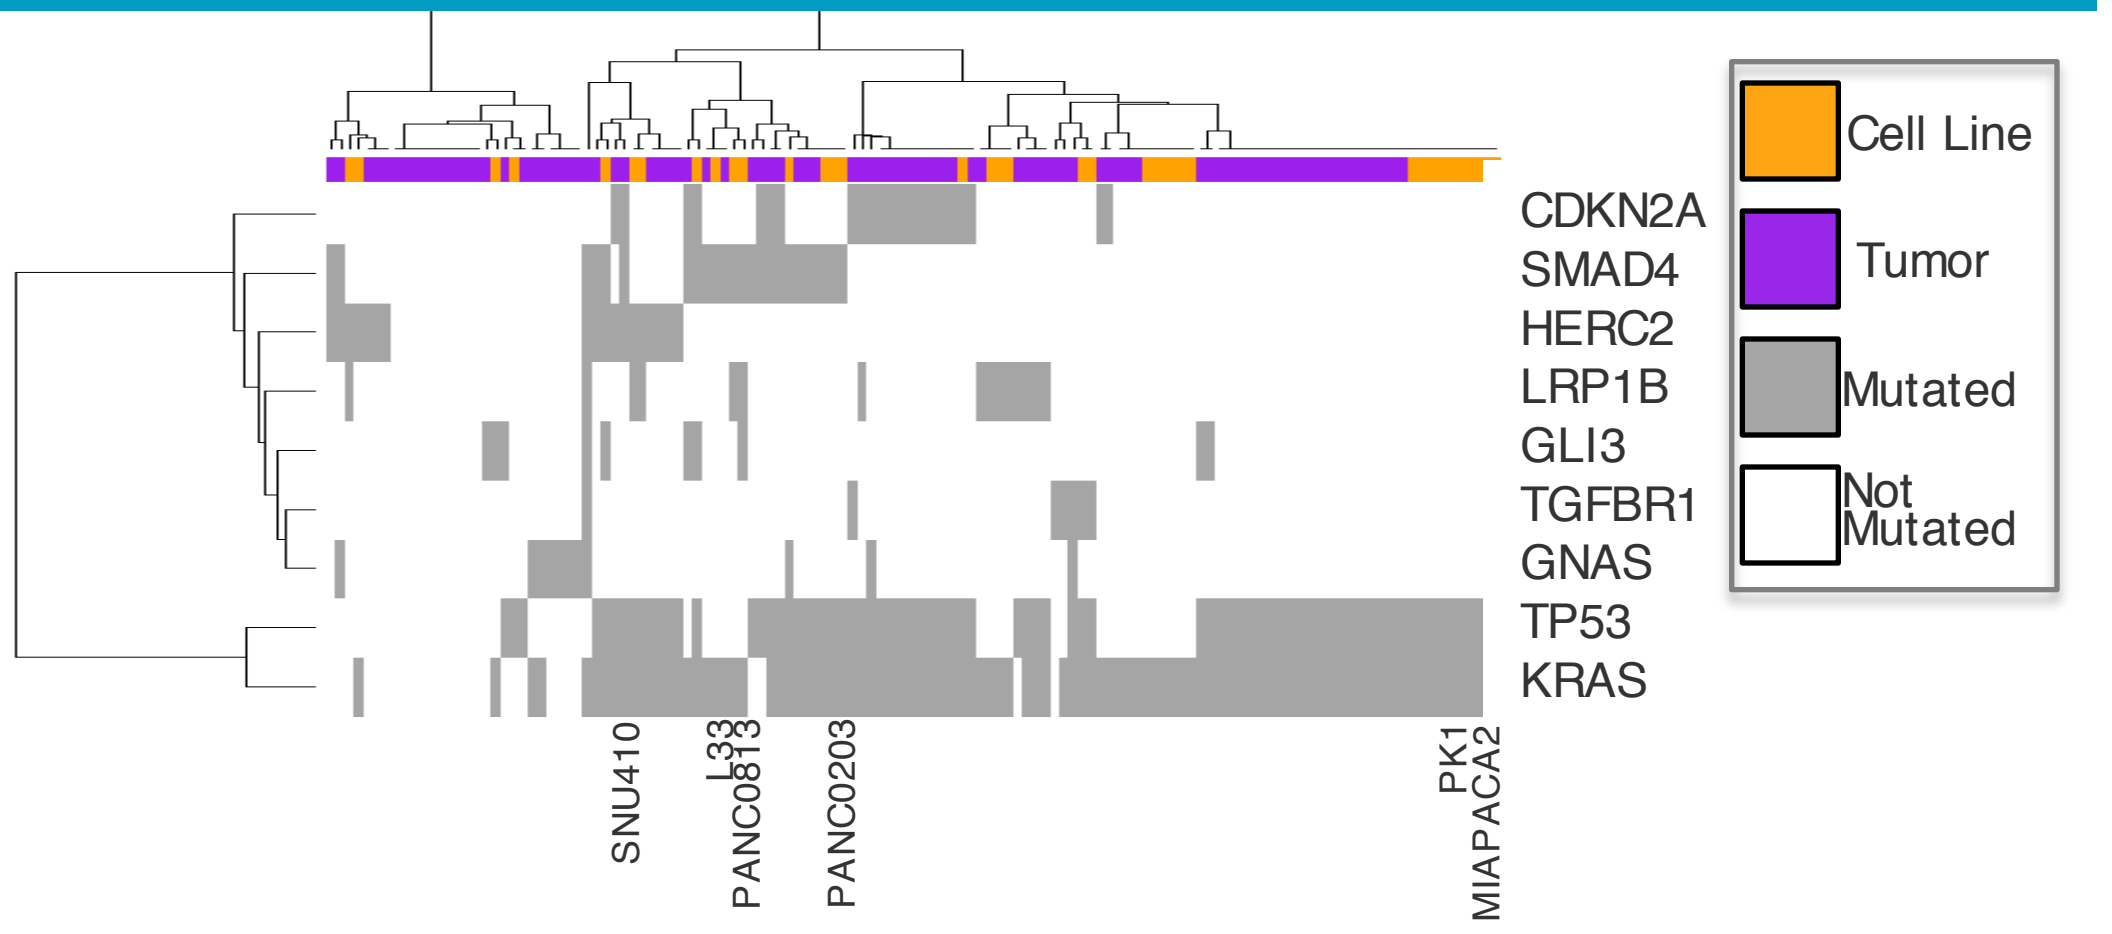
Cell Line
Tumor
Mutated
Not Mutated
CDKN2A
SMAD4
HERC2
LRP1B
GLI3
TGFBR1
GNAS
TP53
KRAS
SNU410
L33
PANC0813
PANC0203
PK1
MIAPACA2

Figure 6. Using hierarchical clustering based on the presence or absence of the mutations passing filter, we showed that some cell lines readily clustered amongst tumors. Interestingly L33 and PANC0203 cluster in a branch with many tumor branches whereas MIAPACA2 occupies a branch tumors and cell lines that only have the TP53 and KRAS mutations.

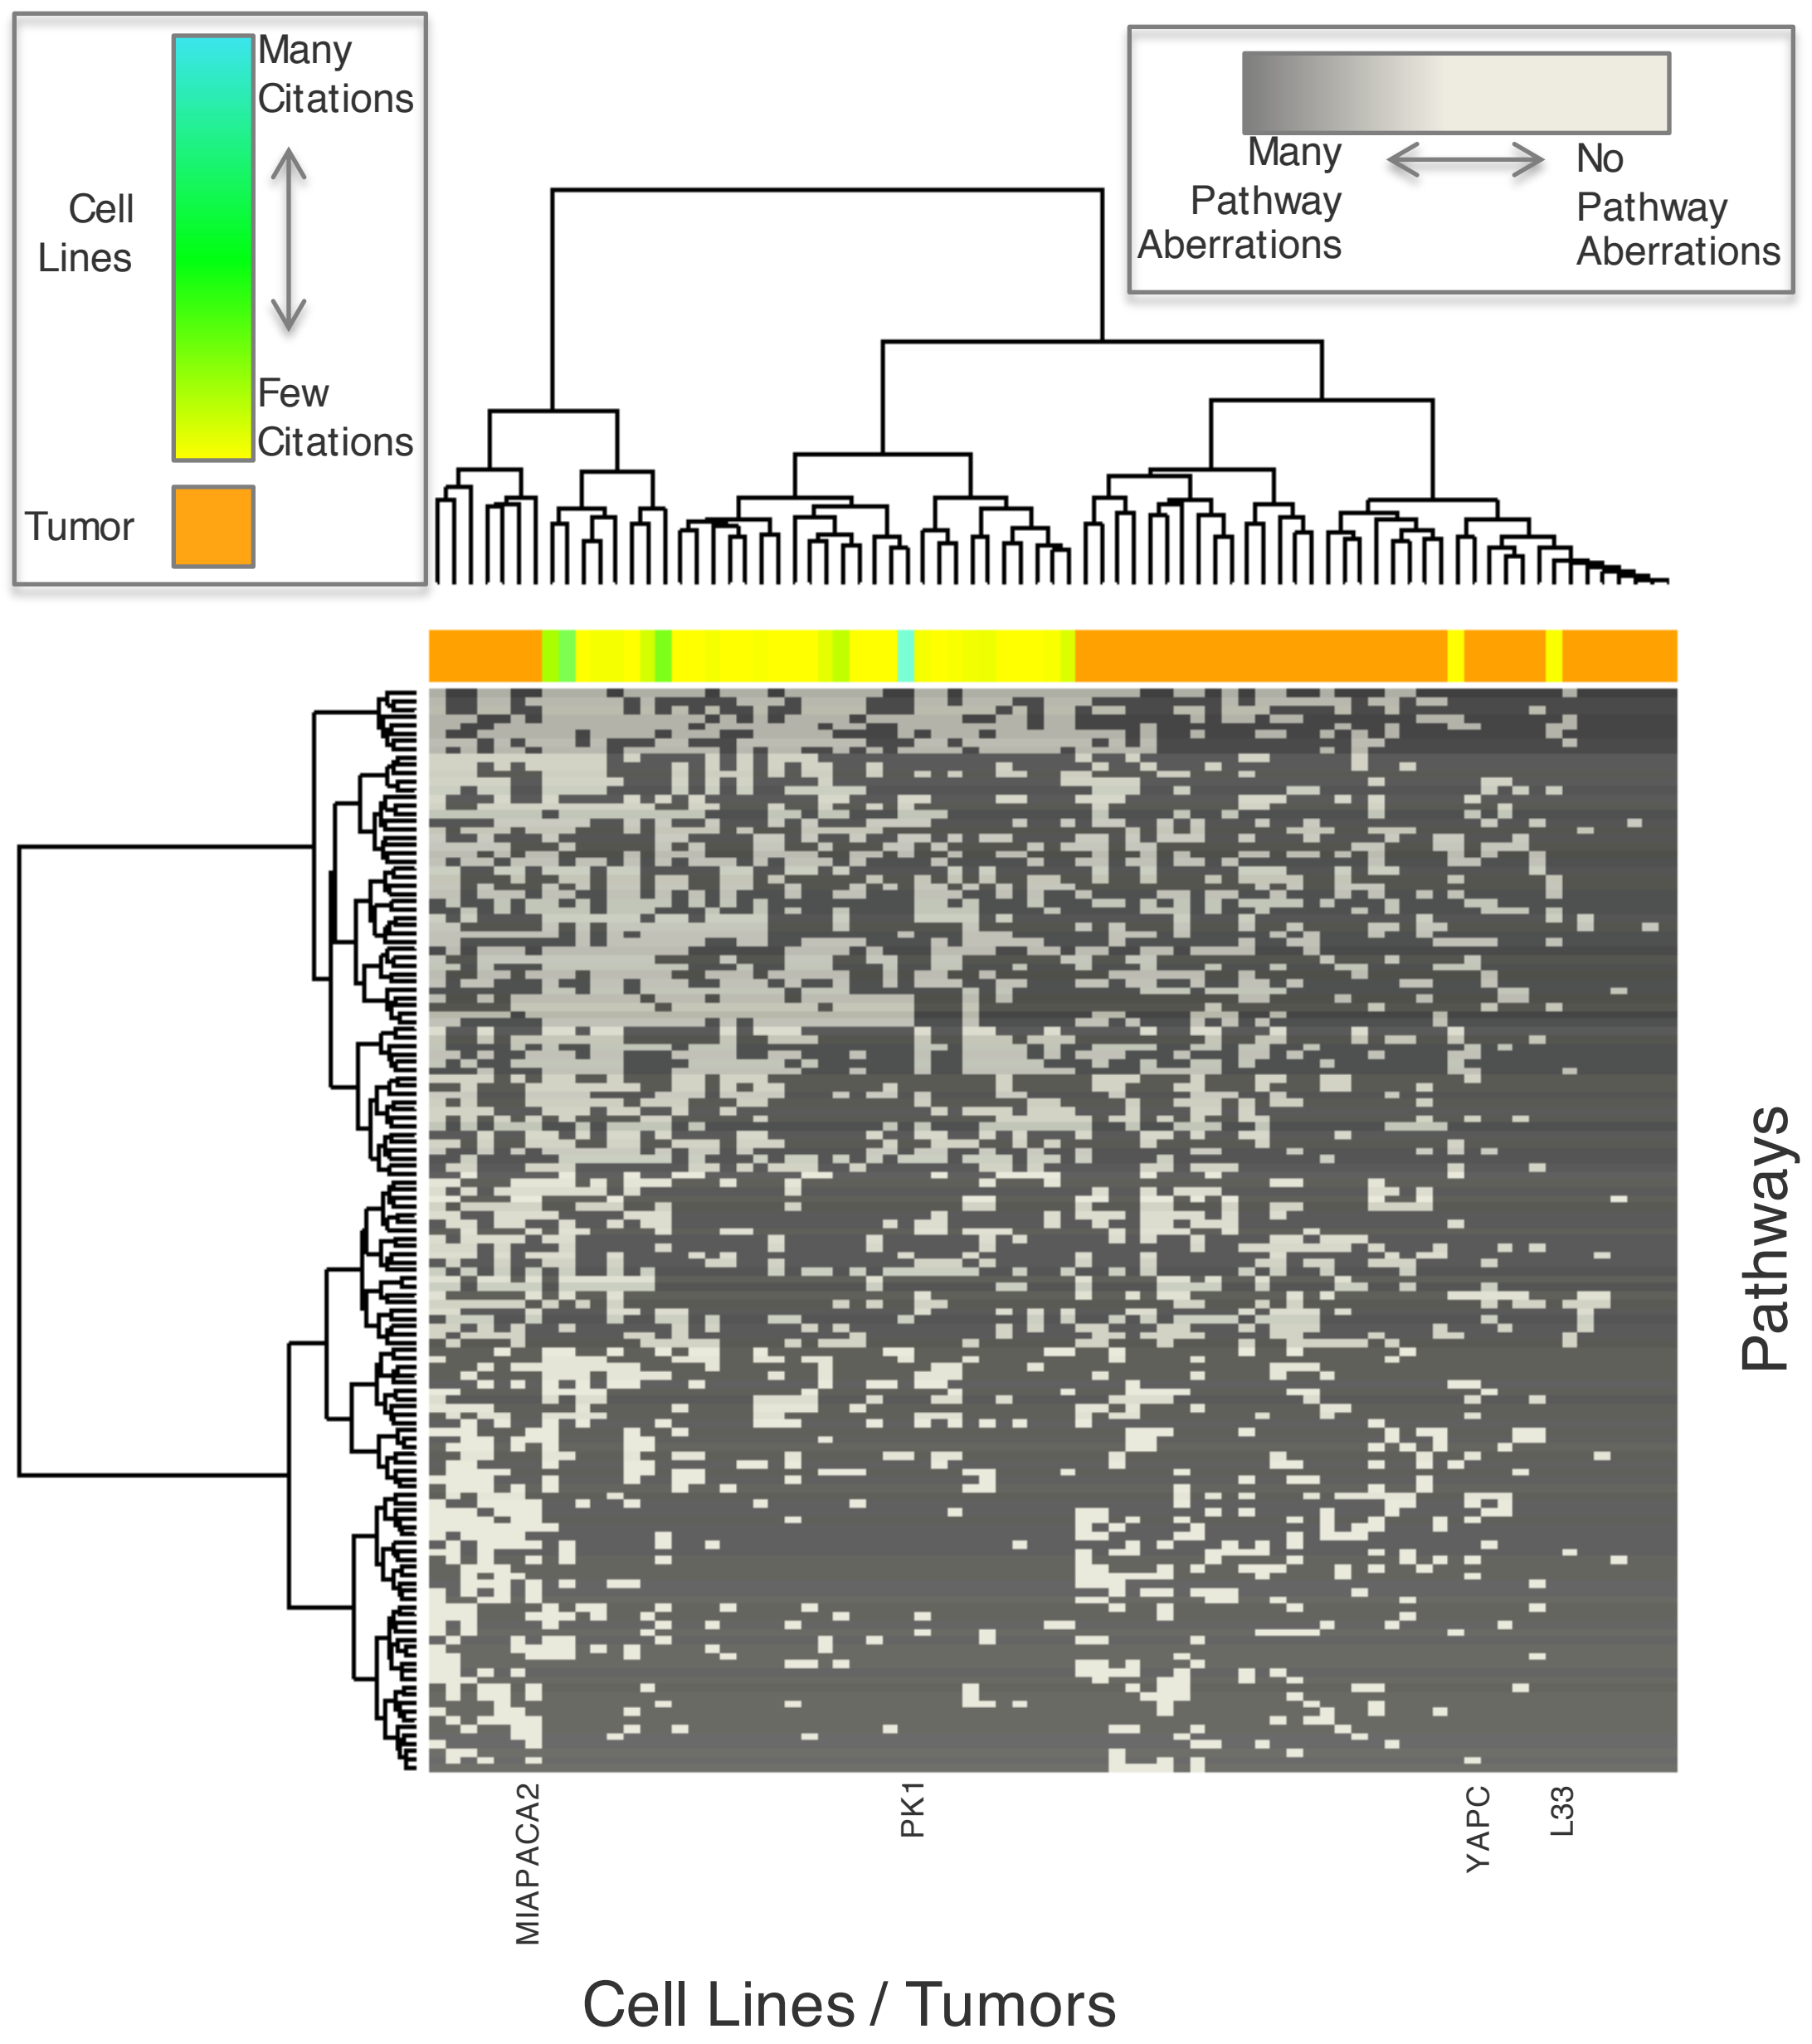
Cell Lines
Many Citations
Few Citations
Tumor
Many Pathway Aberrations
No Pathway Aberrations
Pathways
MIAPACA2
PK1
YAPC
L33
Cell Lines / Tumors

Figure 7. Select cell lines cluster with tumors. Interestingly L33 and YAPC cluster with pancreatic cancer tumors. All other cell lines cluster in branches composed of other cell lines. This shows that many popular cell lines may have genomic differences from tumors in important pathways.

Conclusions

- Based on the present analysis, some cell lines used less frequently in literature such as L33, SNU410 and PANC0203, may more accurately represent tumors as compared to popular cell lines MIAPACA2.
- Our work reports that many popular pancreatic cancer cell lines harbor distinct genomic aberration profiles from pancreatic cancer tumors and highlights the emerging role of genomics in advancing the clinical success of therapeutic trials buy accelerate pre-clinical drug screening.

Future Directions

- It is possible to apply this method to other cancer types, given consideration for potentially different cancer biology.
- Drug response across patients is often heterogeneous, thus future methods will take genomic tumor subclass into account.

Immuneering
Contact: Rebecca Kusko (bkusko@immuneering.com, 617-500-8080 ext. 121, www.immuneering.com), Yoonjeong Cha (ycha@immueering.com)