Preliminary Phase 1 Safety and Activity of IMM-1-104, an Orally Dosed Universal RAS Inhibitor That Drives Deep Cyclic Inhibition of the MAPK Pathway at MEK, in Patients with Advanced Unresectable or Metastatic Solid Tumors

V. Chung[1], A. Spira[2], A. Pavlick[3], D. Sommerhalder[4], B. Ma[3], V. Hayreh[5], J. de Jong[5], J. Funt[5], S. Kolitz[5], P. Nair[5], J. Zhang[5], J. Kim[5], A. Yamamura[5], B. Zeskind[5], B. Hall[5], S. Pant[6]

[1]City of Hope, Duarte, CA, [2]NEXT, VA, [3]Cornell, New York, NY, [4]NEXT, San Antonio, TX, [5]Sponsor: Immuneering Corporation, Cambridge, MA, [6]MD Anderson Cancer Center, Houston, TX

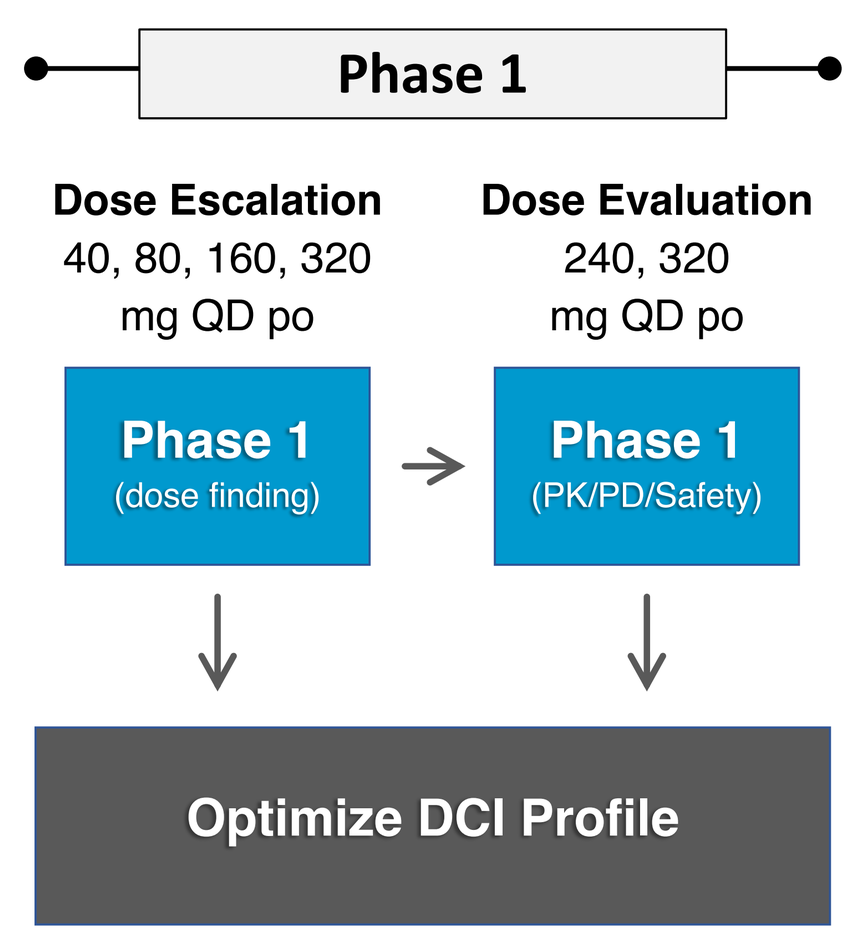
Phase 1
Dose Escalation
40, 80, 160, 320 mg QD po
Dose Evaluation
240, 320 mg QD po
Phase 1
(dose finding)
Phase 1
(PK/PD/Safety)
Optimize DCI Profile

Background: About 33% of tumors have activating RAS mutations making it an attractive target. MEK inhibitors block the downstream signaling of RAS, but earlier inhibitors have been plagued with toxicities. IMM-1-104, a novel MEK inhibitor, blocks both MEK and ERK phosphorylation preventing CRAF bypass and hyperactivation of MEK. A short half-life of 2 hours combined with once daily dosing (QD) drives deep, pulsatile inhibition of the MAPK pathway at MEK in a process called Deep Cyclic Inhibition (DCI). This improves drug safety and tolerability by allowing daily pathway recovery in healthy tissues while limiting adaptive resistance in tumor cells[1].

Methods: We conducted a first-in-human phase 1 study of IMM-1-104 at 5 U.S. sites. The primary objective was to determine the safety, tolerability and recommended phase 2 dose of IMM-1-104 in advanced, RAS-mutated solid tumors. A rapid dose escalation scheme was used with broad inclusion criteria, and PK, PD (circulating tumor DNA (ctDNA) and *ex vivo* pERK) and initial clinical efficacy were evaluated.

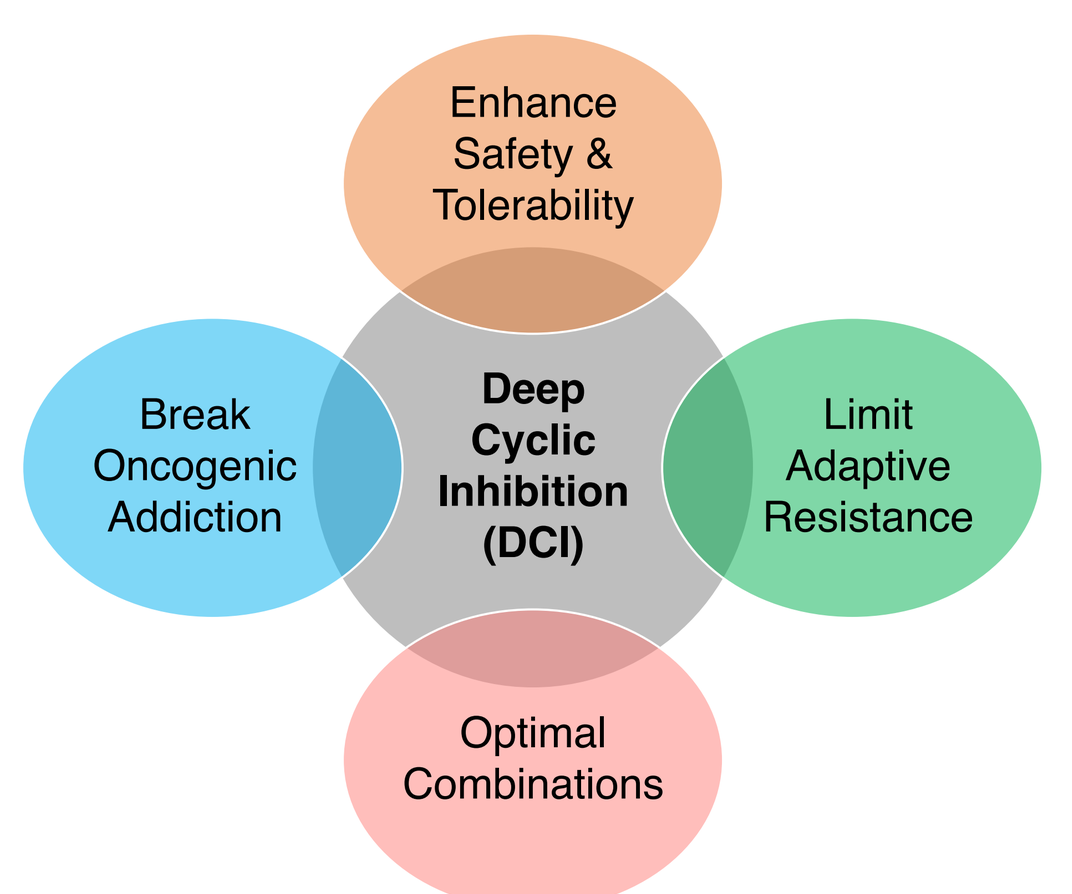
Enhance Safety & Tolerability
Break Oncogenic Addiction
Deep Cyclic Inhibition (DCI)
Limit Adaptive Resistance
Optimal Combinations

Results: As of August 19, 2024, the phase 1 study enrolled 54 subjects, including 34 PDAC, 8 colorectal, 5 lung, 2 melanoma, 2 appendiceal, 1 cholangiocarcinoma plus 2 others. Evaluation of two candidate optimal doses, 240 and 320 mg QD, supported DCI of the MAPK pathway. Treatment-related adverse events (TRAEs) occurring in ≥10% of patients were transient and limited mainly to grade 1 or 2, when observed. No DLTs or serious TRAEs were noted. Although RECIST responses were not observed nor an endpoint of Phase 1, promising signs of clinical activity in heterogeneous, heavily pretreated, advanced metastatic patients were evident. Paired CT scans (40) and ctDNA (36) revealed: (1.) 27 (68%) had SLDs < 20%, 9 (23%) had SLDs 0% to -25%, (2.) 15 (38%) had ≥ 1 target lesion regression (-2.0% to -66.7%), (3.) mean ctDNA reductions were observed in 13 (36%) patients (-3% to -81%), (4.) no new mutation variants in RAS ctDNA and (5.) dose-dependent inhibition *ex vivo* pERK[1].

IMM-1-104 Phase 1 Data Show Promising Safety, Tolerability and Early Monotherapy Activity – 4 Highlighted Case Studies

Patient 1: KRAS^{G12V} PDAC

- Metastatic Pancreatic Cancer (adeno)
- 66-year-old Hispanic male
- 6 Prior lines of treatment (BoR = SD)
- 6th Line: IFL (BoR = SD)
- 7th Line: **240 mg QD p.o. IMM-1-104**
- IMM-1-104 BoR = **Stable Disease**
- 97 days on study drug (lesions expanding)

Patient 2: KRAS^{G12D} PDAC

- Metastatic Pancreatic Cancer (adeno)
- 74-year-old Asian male
- 1st Line: FOLFIRINOX (BoR = PR)
- 2nd Line: **320 mg QD p.o. IMM-1-104**
- IMM-1-104 BoR = **Progressive Disease**
- 55 days on study drug (mixed response)
- NRAS mutation identified and eliminated

Patient 3: KRAS^{G12D} PDAC

- Metastatic Pancreatic Cancer (adeno)
- 64-year-old Caucasian male
- 1st Line: FOLFIRINOX (BoR = SD)
- 2nd Line: **320 mg QD p.o. IMM-1-104**
- IMM-1-104 BoR = **Stable Disease**
- 84 days on study drug (lesions regressing)
- 6-8 hours of daily pain relief

Patient 4: KRAS^{G12D} PDAC

- Metastatic Pancreatic Cancer (adeno)
- 70-year-old Caucasian male
- 1st Line: FOLFIRINOX (BoR = PD)
- 2nd Line: Gem, Cis, nab-Pac (BoR = PD)
- 3rd Line: **240 mg QD p.o. IMM-1-104**
- IMM-1-104 (on study) = ***Stable Disease**
- Improved QoL[2] and weight gain (+7%)

RECIST Target Lesions (SLD)

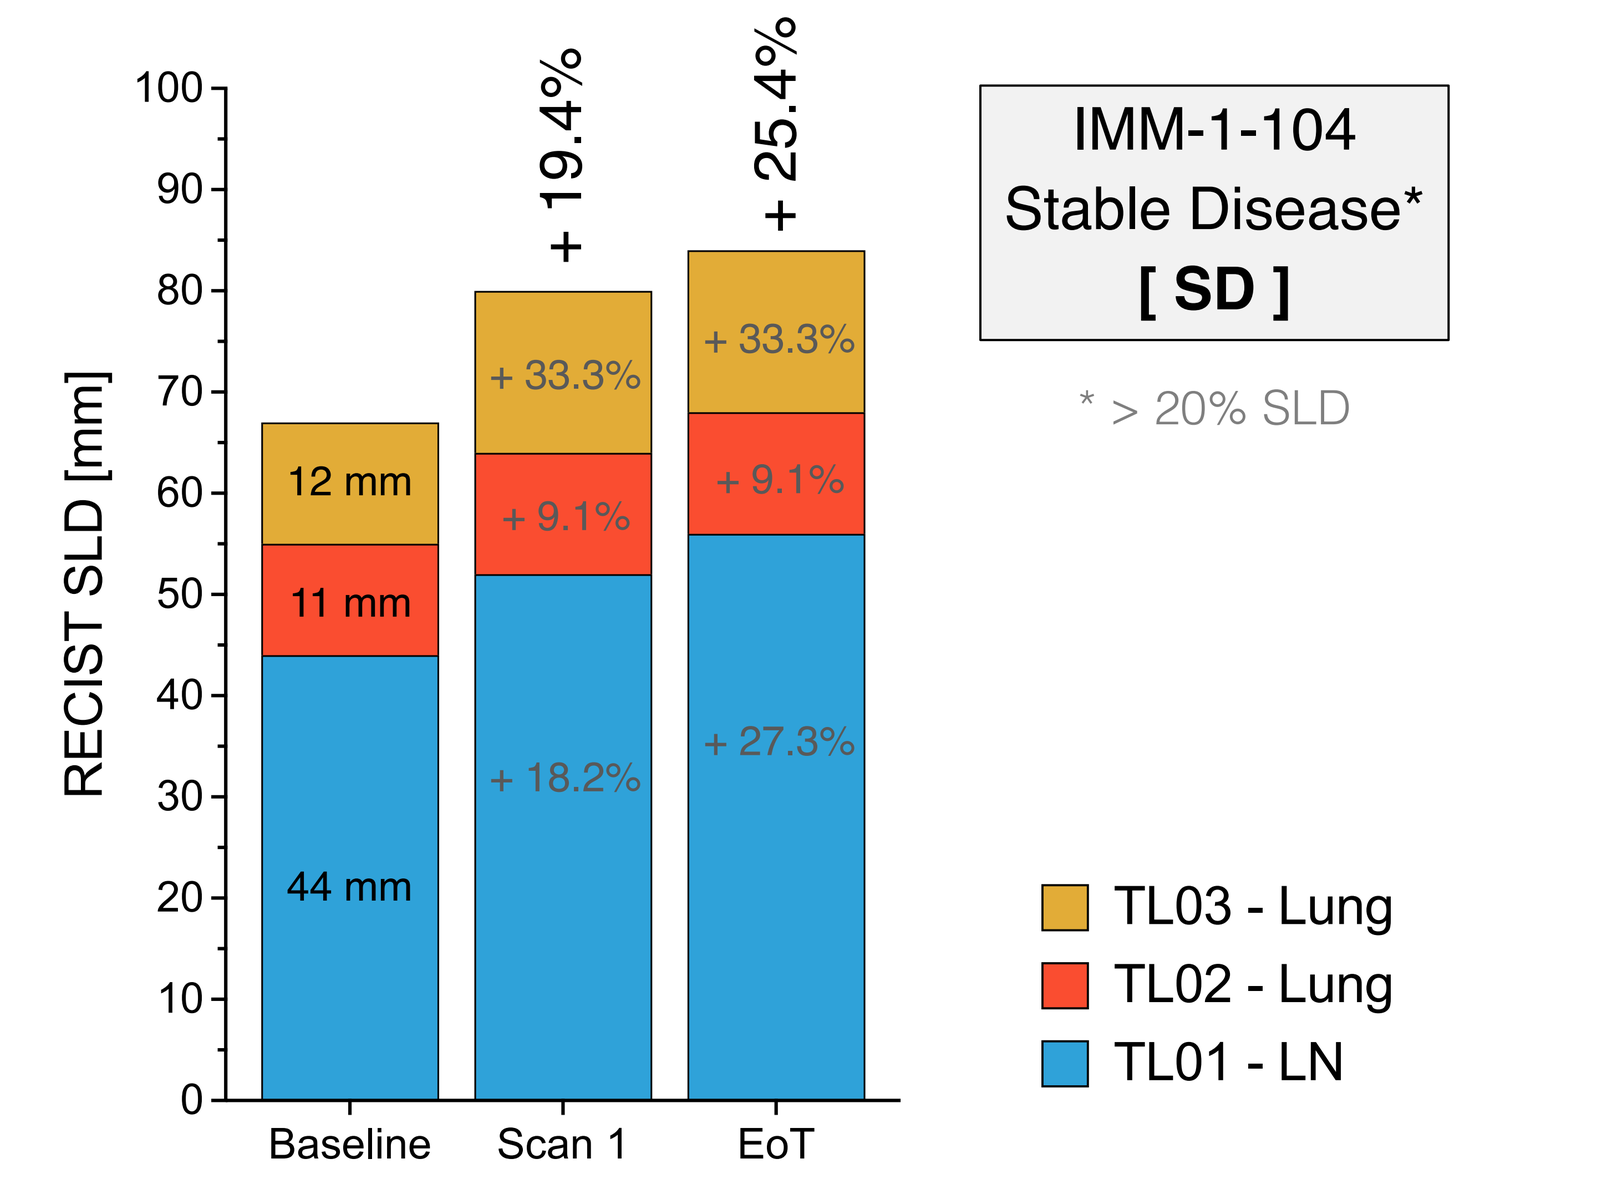
RECIST SLD [mm]
+ 19.4%
+ 25.4%
12 mm
11 mm
44 mm
+ 33.3%
+ 9.1%
+ 18.2%
+ 33.3%
+ 9.1%
+ 27.3%
Baseline
Scan 1
EoT
IMM-1-104
Stable Disease*
[SD]
* > 20% SLD
TL03 - Lung
TL02 - Lung
TL01 - LN

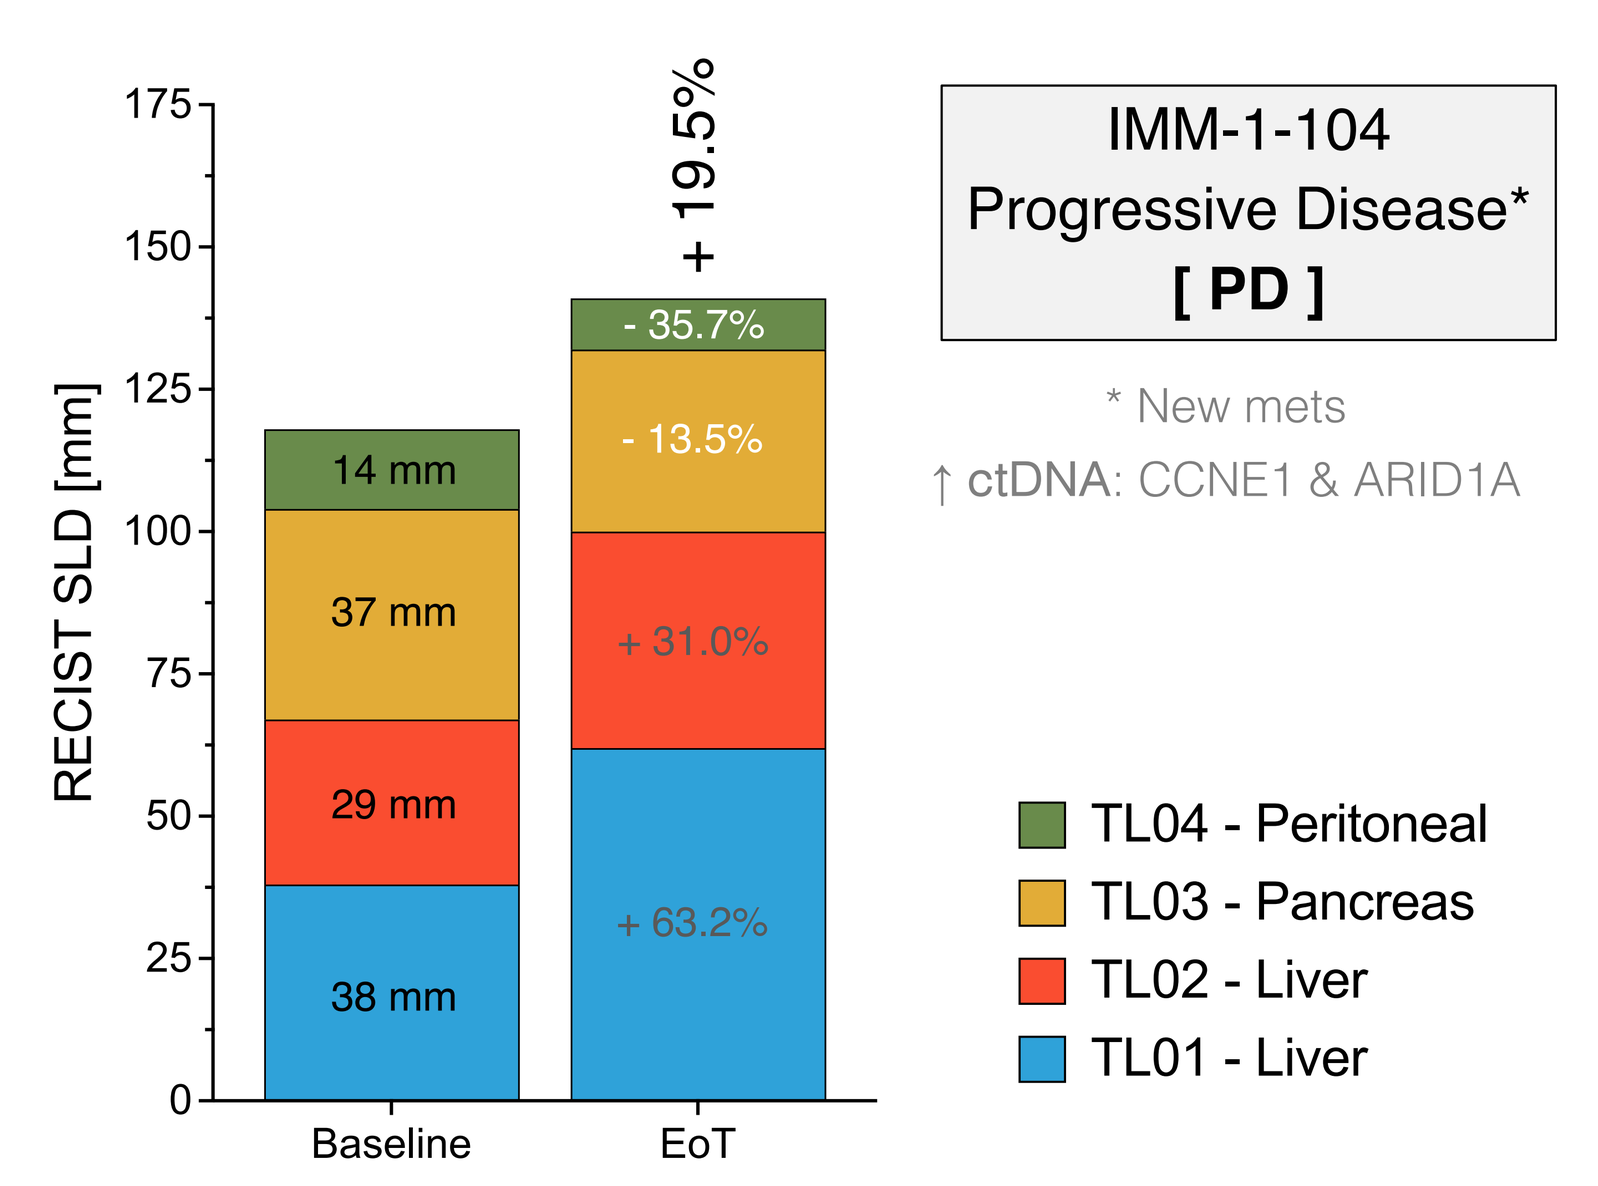
RECIST SLD [mm]
+ 19.5%
14 mm
37 mm
29 mm
38 mm
- 35.7%
- 13.5%
+ 31.0%
+ 63.2%
Baseline
EoT
IMM-1-104
Progressive Disease*
[PD]
* New mets
↑ ctDNA: CCNE1 & ARID1A
TL04 - Peritoneal
TL03 - Pancreas
TL02 - Liver
TL01 - Liver

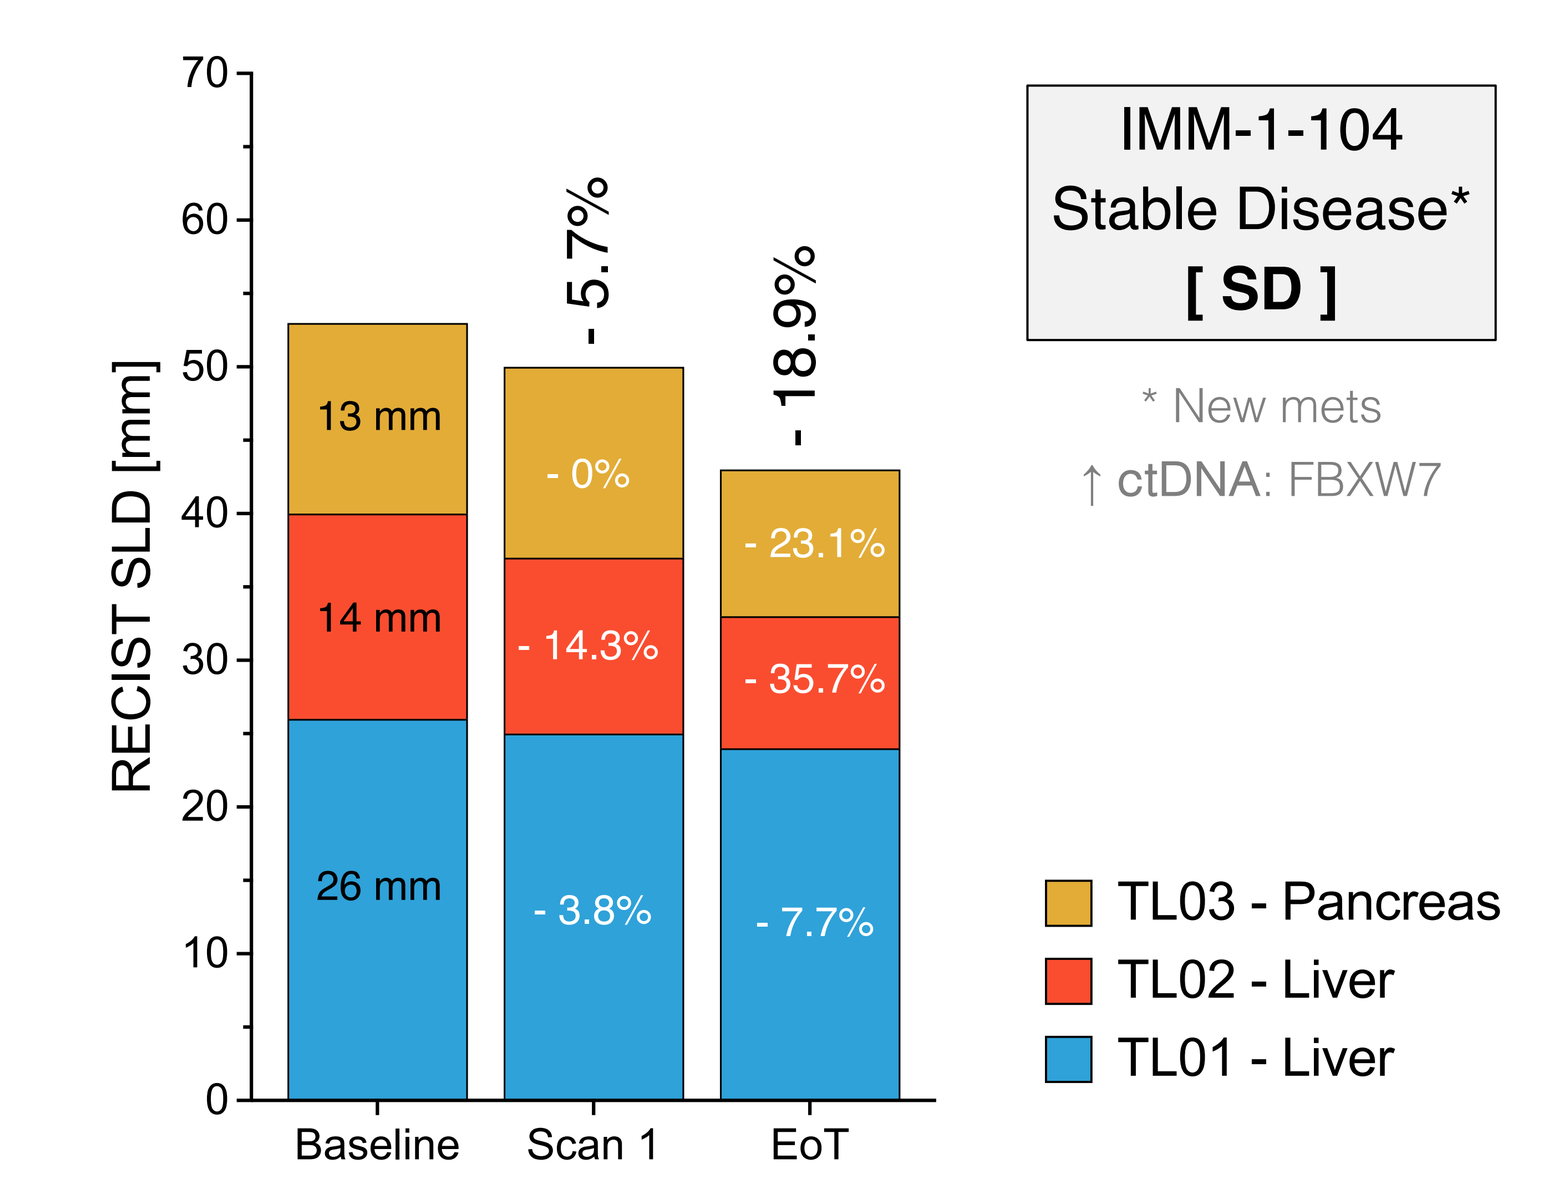
RECIST SLD [mm]
- 5.7%
- 18.9%
13 mm
14 mm
26 mm
- 0%
- 14.3%
- 3.8%
- 23.1%
- 35.7%
- 7.7%
Baseline
Scan 1
EoT
IMM-1-104
Stable Disease*
[SD]
* New mets
↑ ctDNA: FBXW7
TL03 - Pancreas
TL02 - Liver
TL01 - Liver

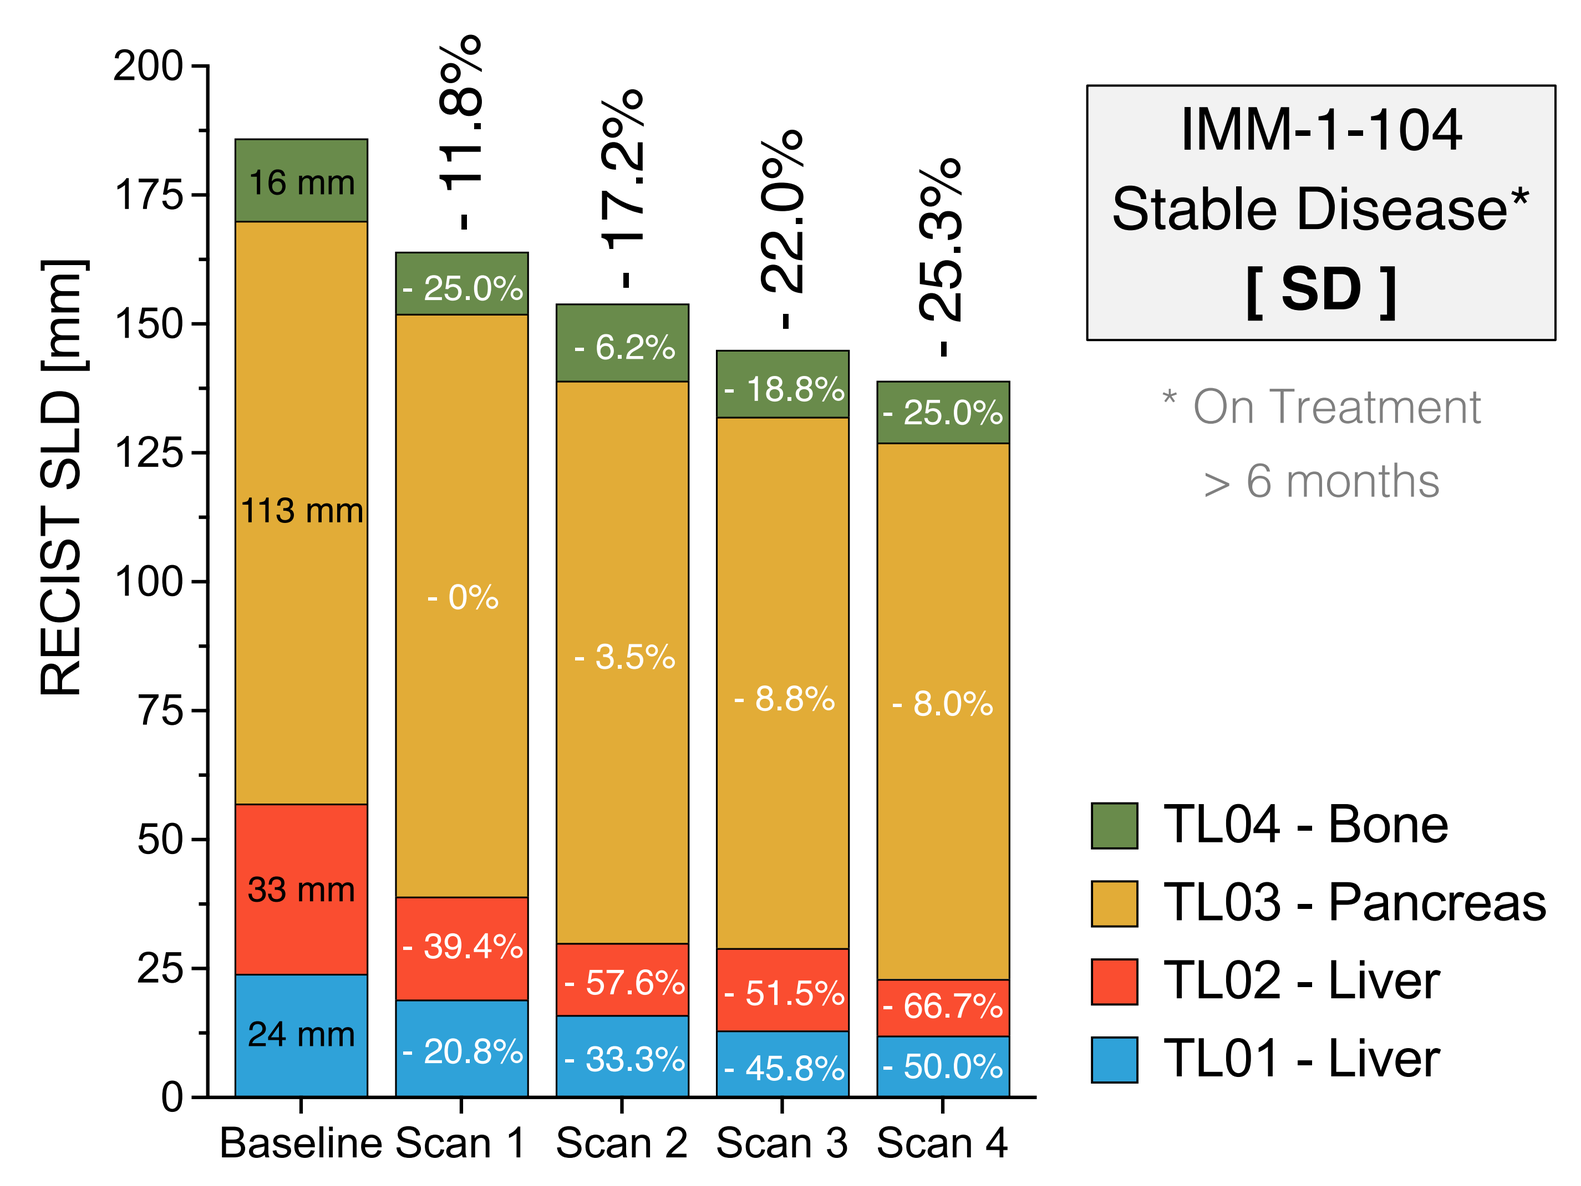
RECIST SLD [mm]
- 11.8%
- 17.2%
- 22.0%
- 25.3%
16 mm
113 mm
33 mm
24 mm
- 25.0%
- 0%
- 39.4%
- 20.8%
- 6.2%
- 3.5%
- 57.6%
- 33.3%
- 18.8%
- 8.8%
- 51.5%
- 45.8%
- 25.0%
- 8.0%
- 66.7%
- 50.0%
Baseline
Scan 1
Scan 2
Scan 3
Scan 4
IMM-1-104
Stable Disease*
[SD]
* On Treatment
> 6 months
TL04 - Bone
TL03 - Pancreas
TL02 - Liver
TL01 - Liver

CA 19-9 (U/mL)

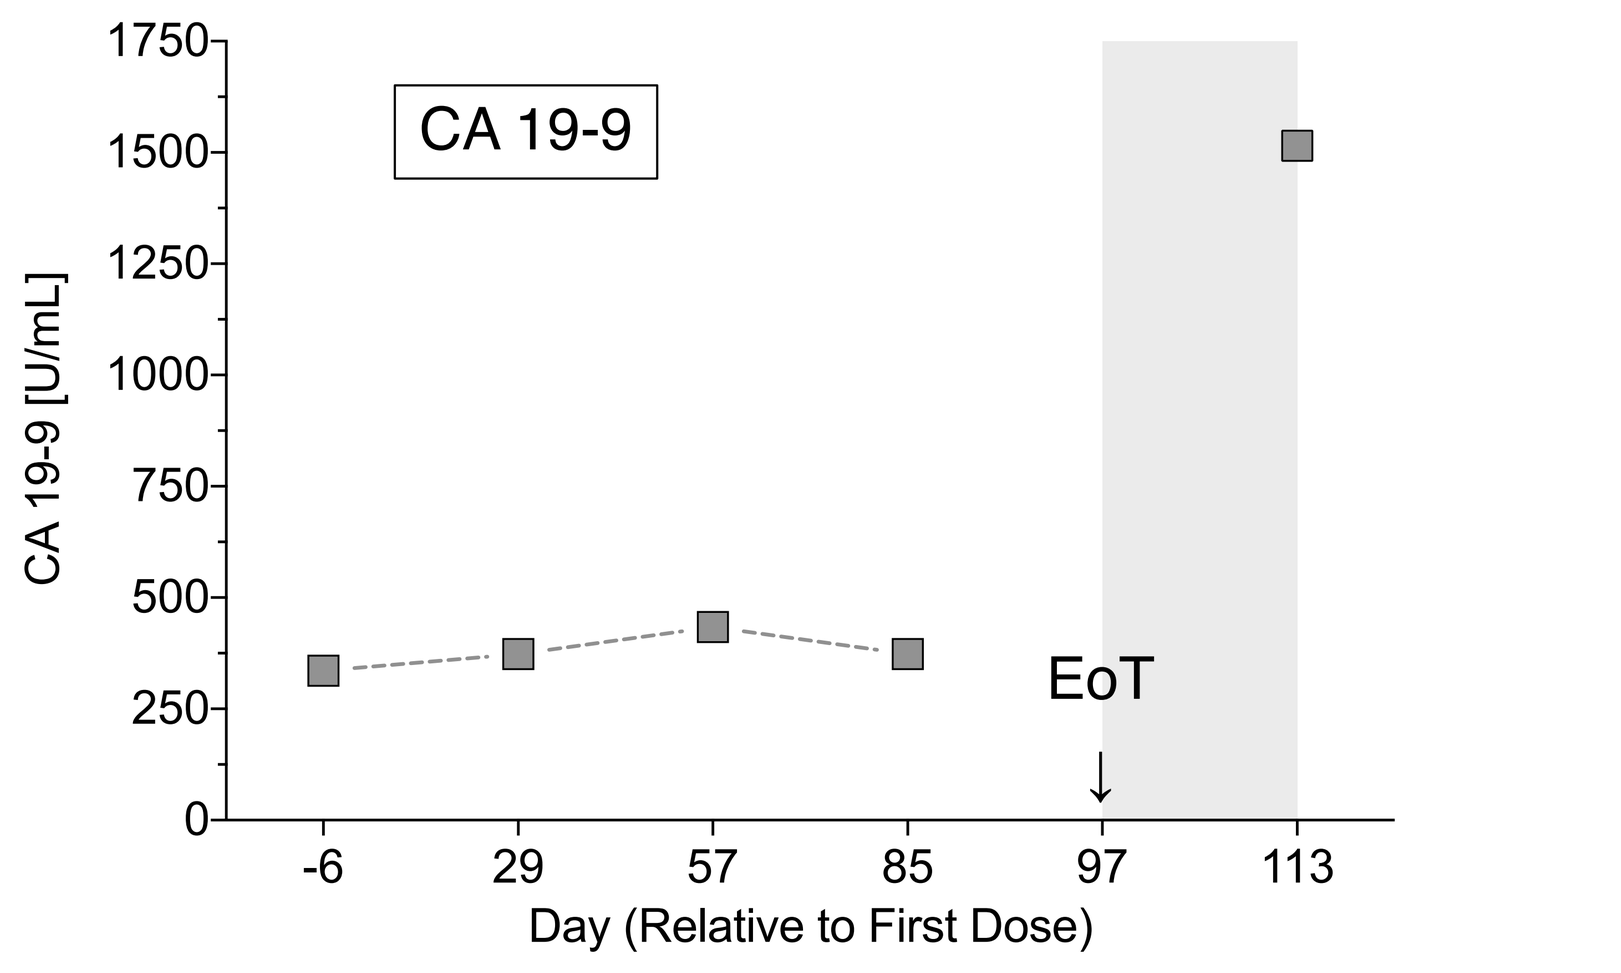
CA 19-9
CA 19-9 [U/mL]
EoT
Day (Relative to First Dose)
-6
29
57
85
97
113

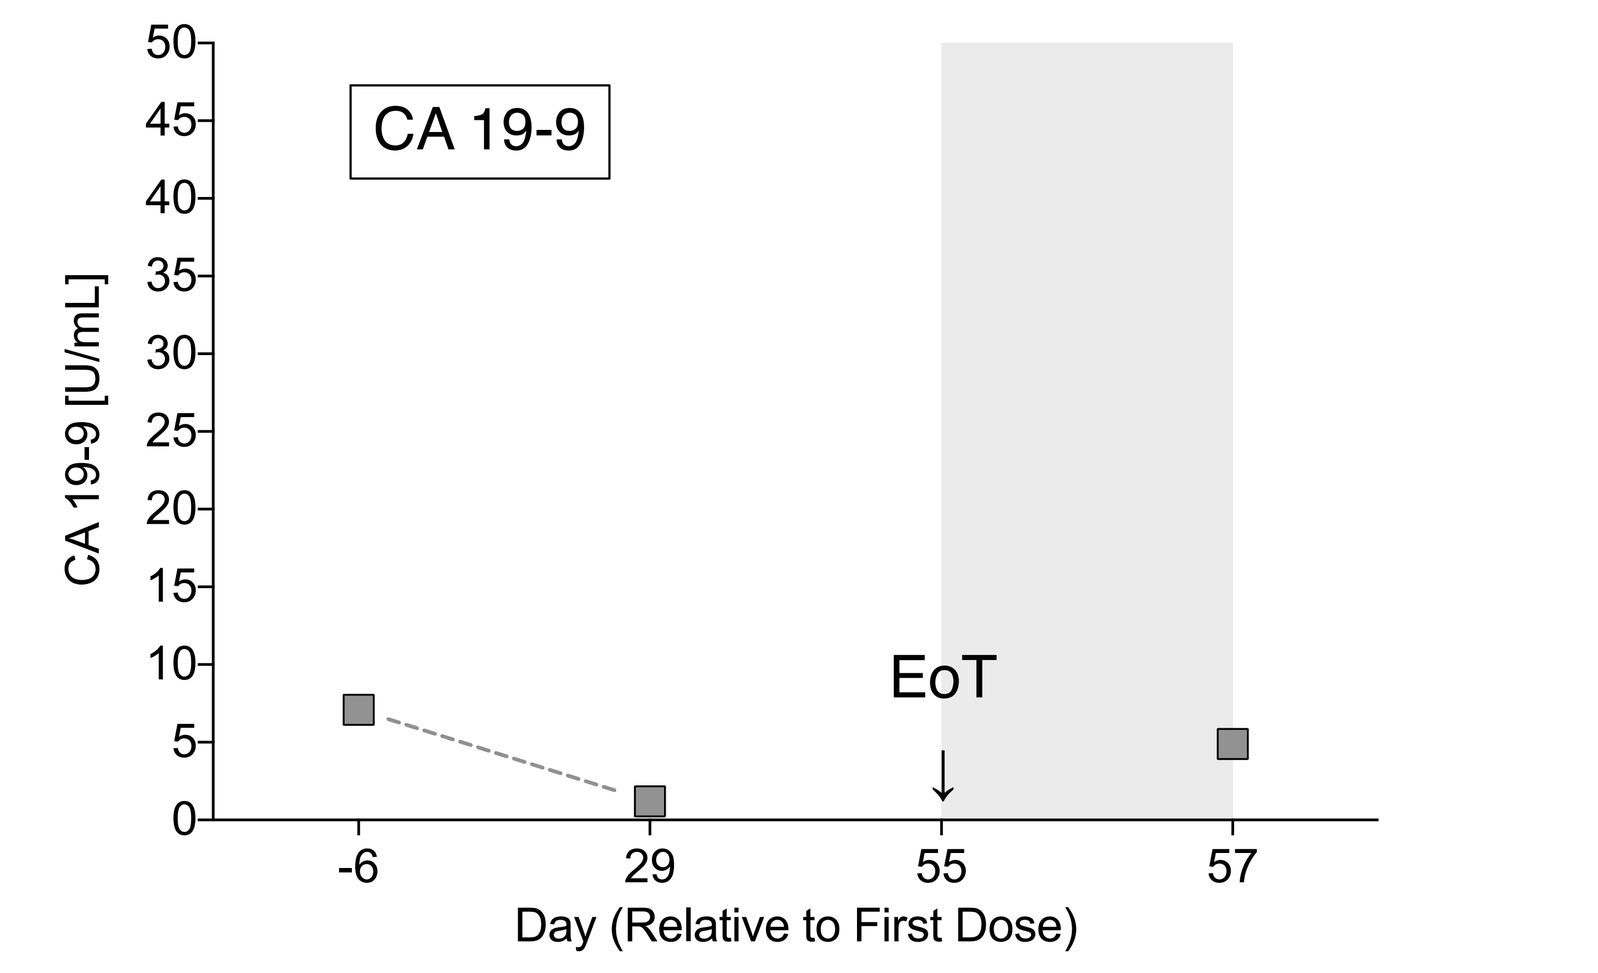
CA 19-9
CA 19-9 [U/mL]
EoT
Day (Relative to First Dose)
-6
29
55
57

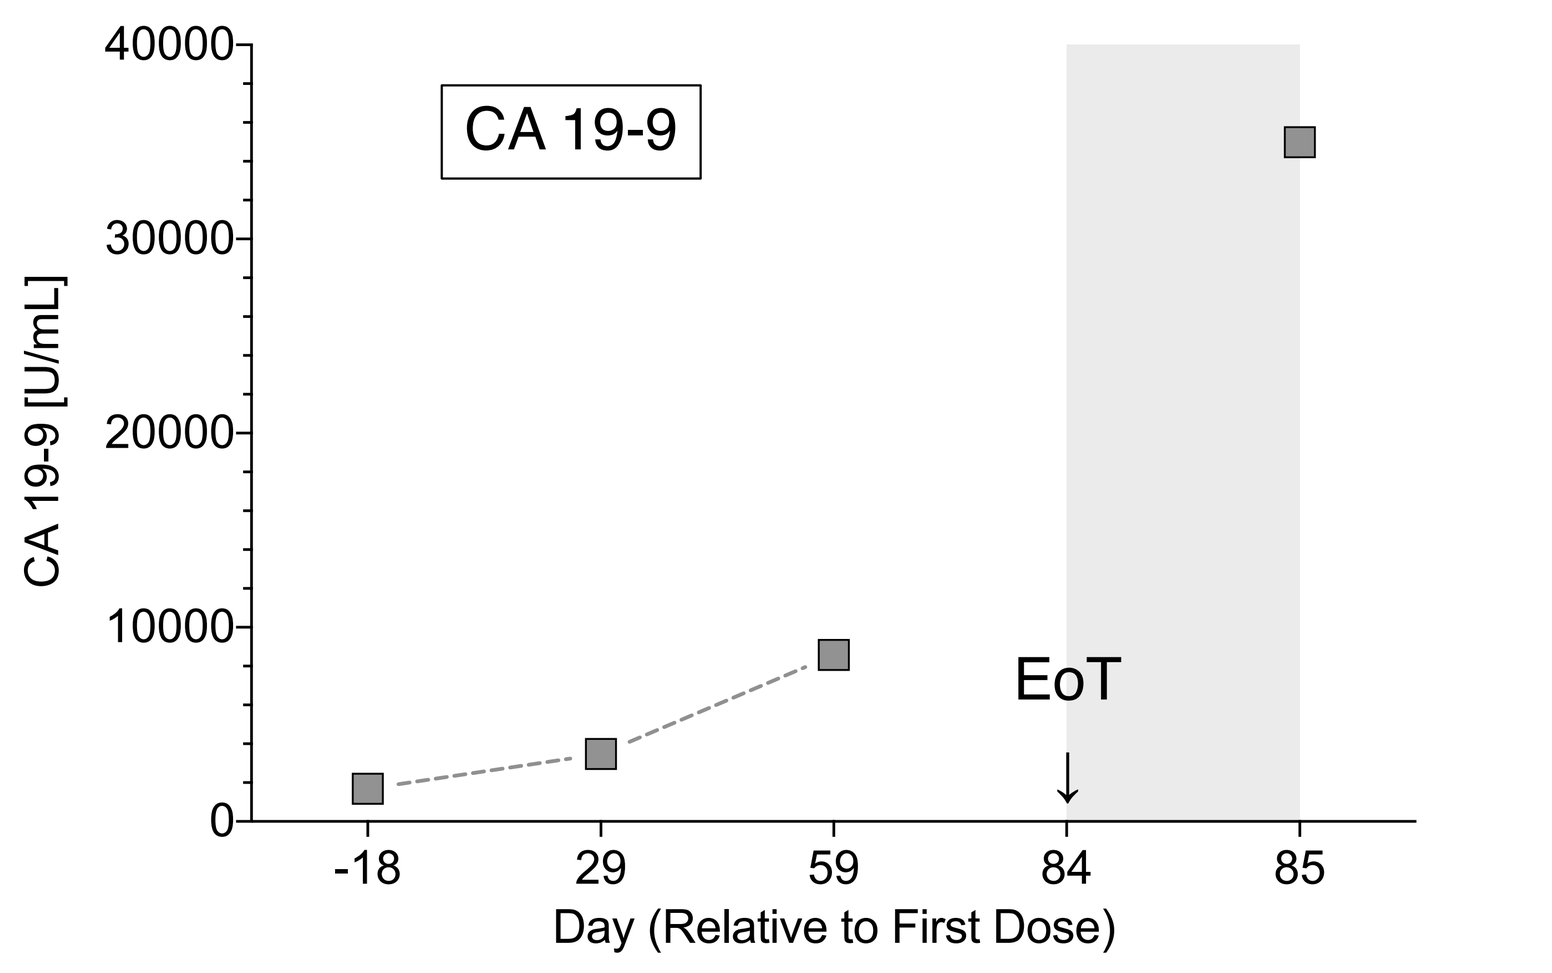
CA 19-9
CA 19-9 [U/mL]
EoT
Day (Relative to First Dose)
-18
29
59
84
85

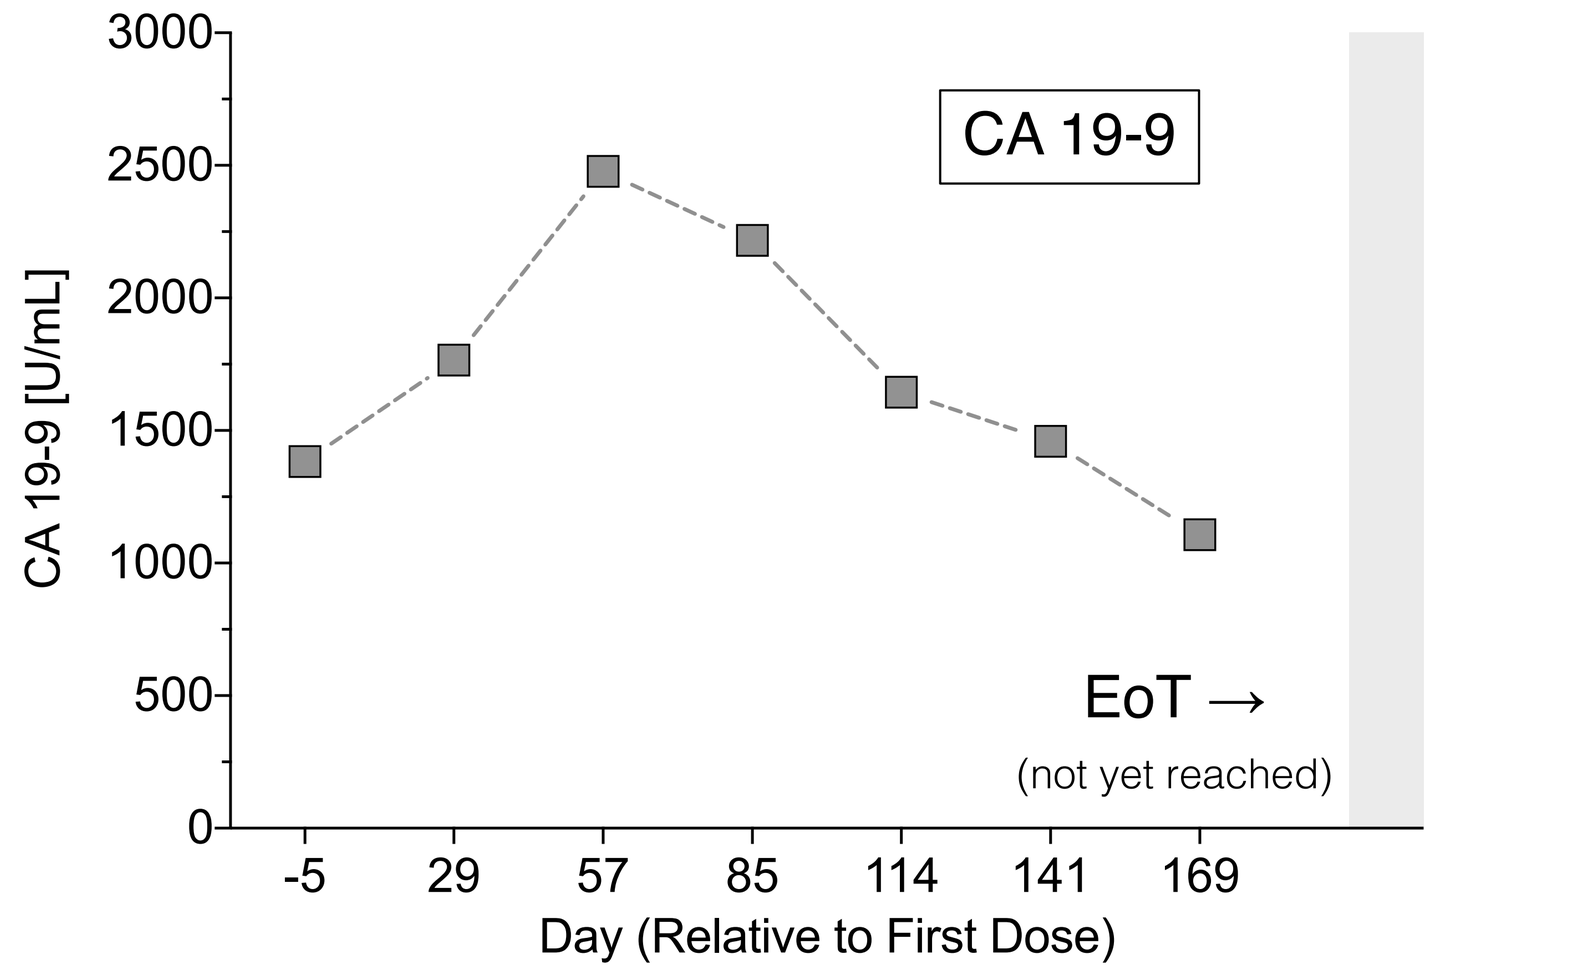
CA 19-9
CA 19-9 [U/mL]
EoT →
(not yet reached)
Day (Relative to First Dose)
-5
29
57
85
114
141
169

ctDNA (VAF)

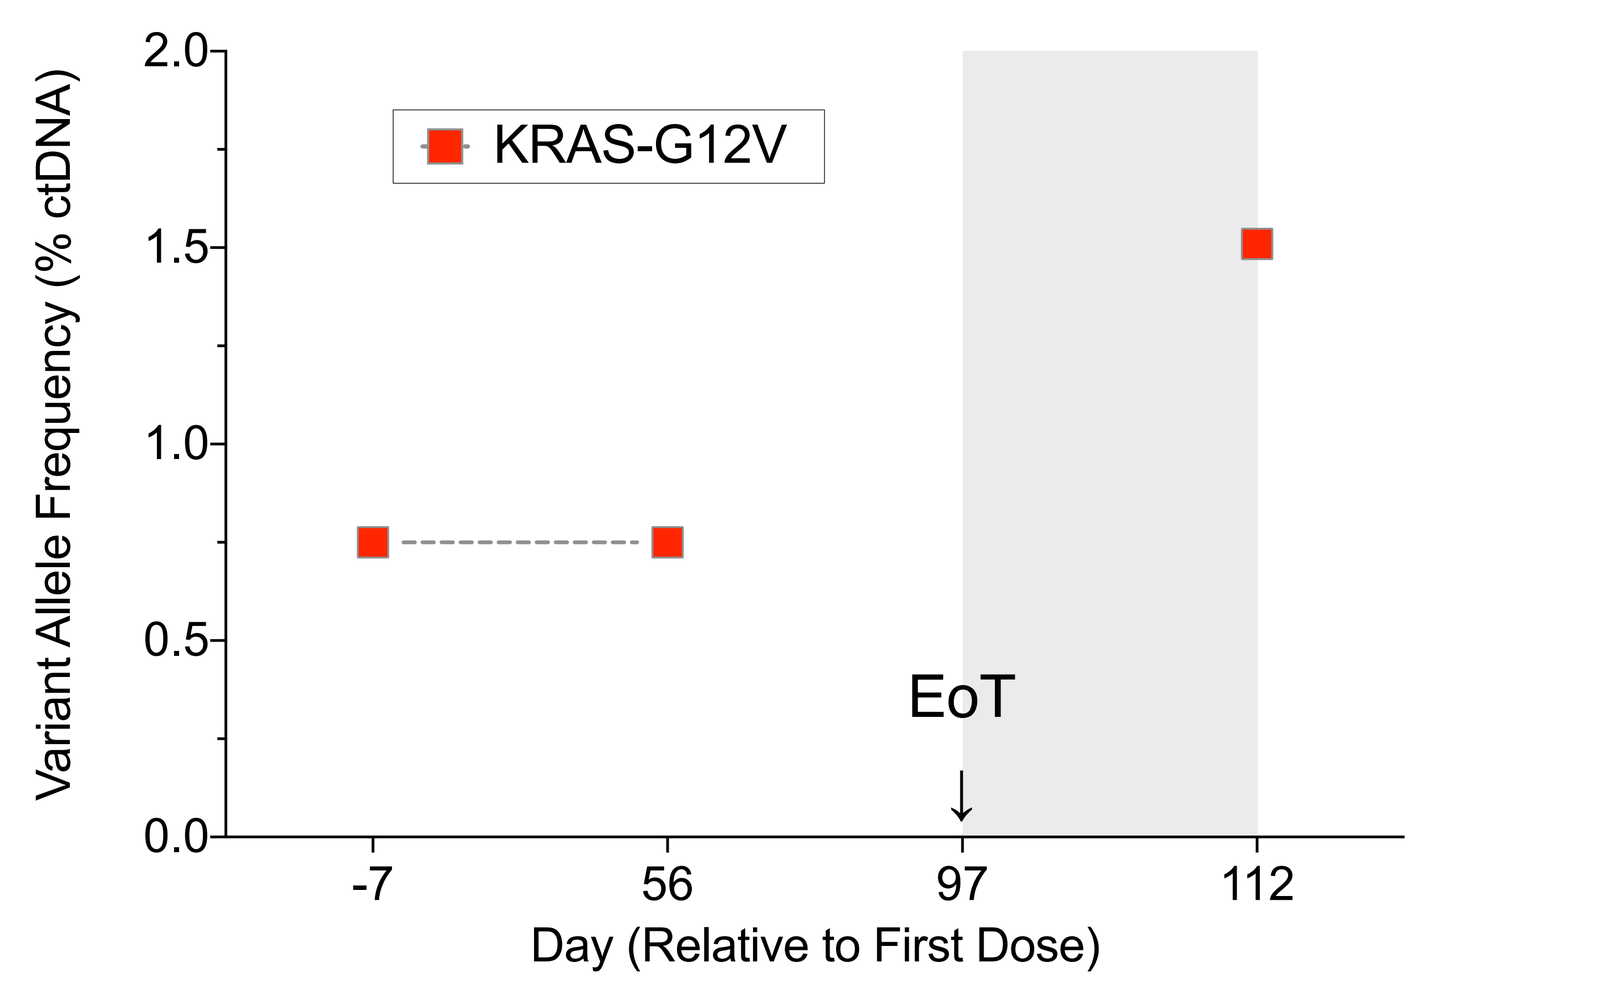
KRAS-G12V
Variant Allele Frequency (% ctDNA)
EoT
Day (Relative to First Dose)
-7
56
97
112

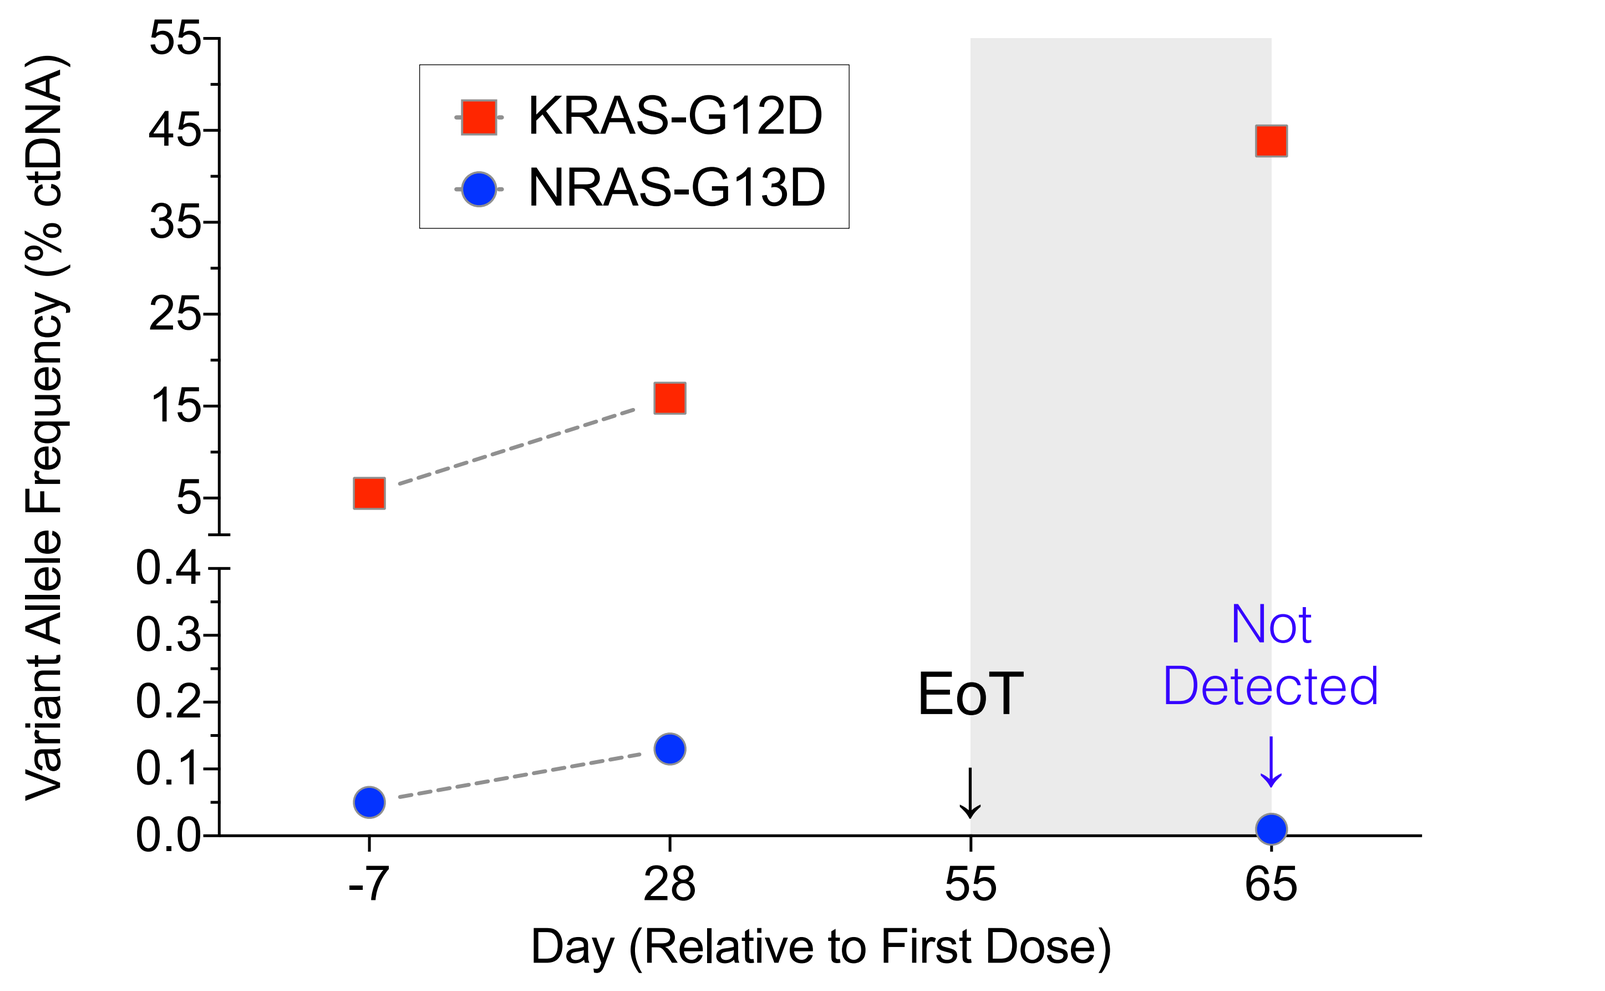
KRAS-G12D
NRAS-G13D
Variant Allele Frequency (% ctDNA)
EoT
Not Detected
Day (Relative to First Dose)
-7
28
55
65

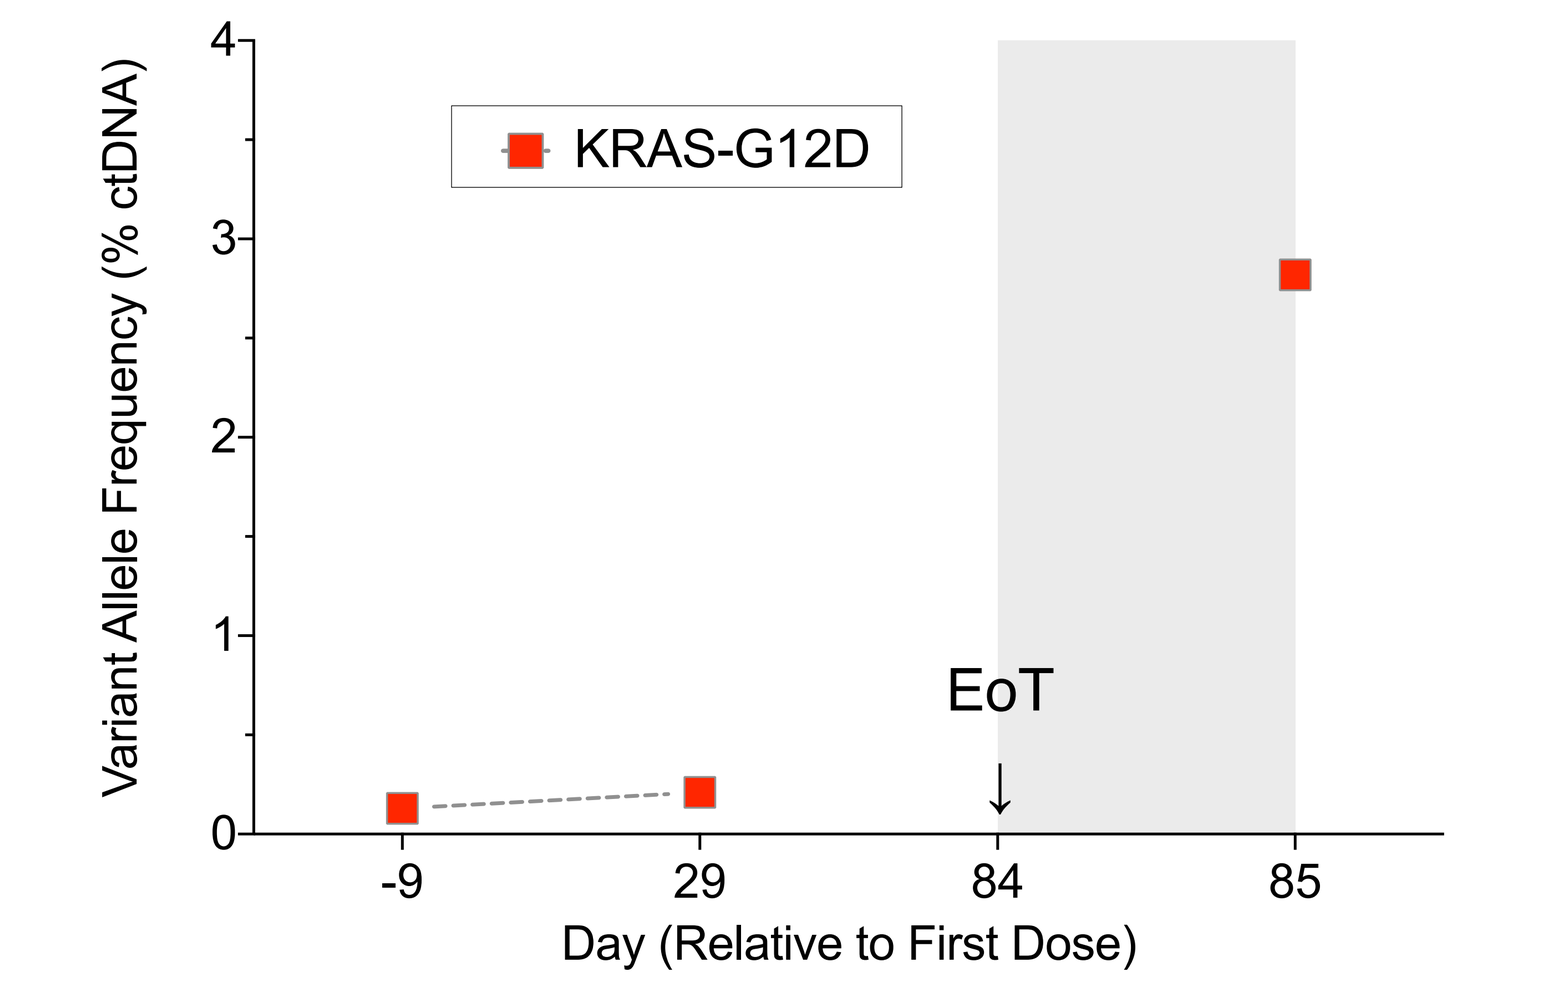
KRAS-G12D
Variant Allele Frequency (% ctDNA)
EoT
Day (Relative to First Dose)
-9
29
84
85

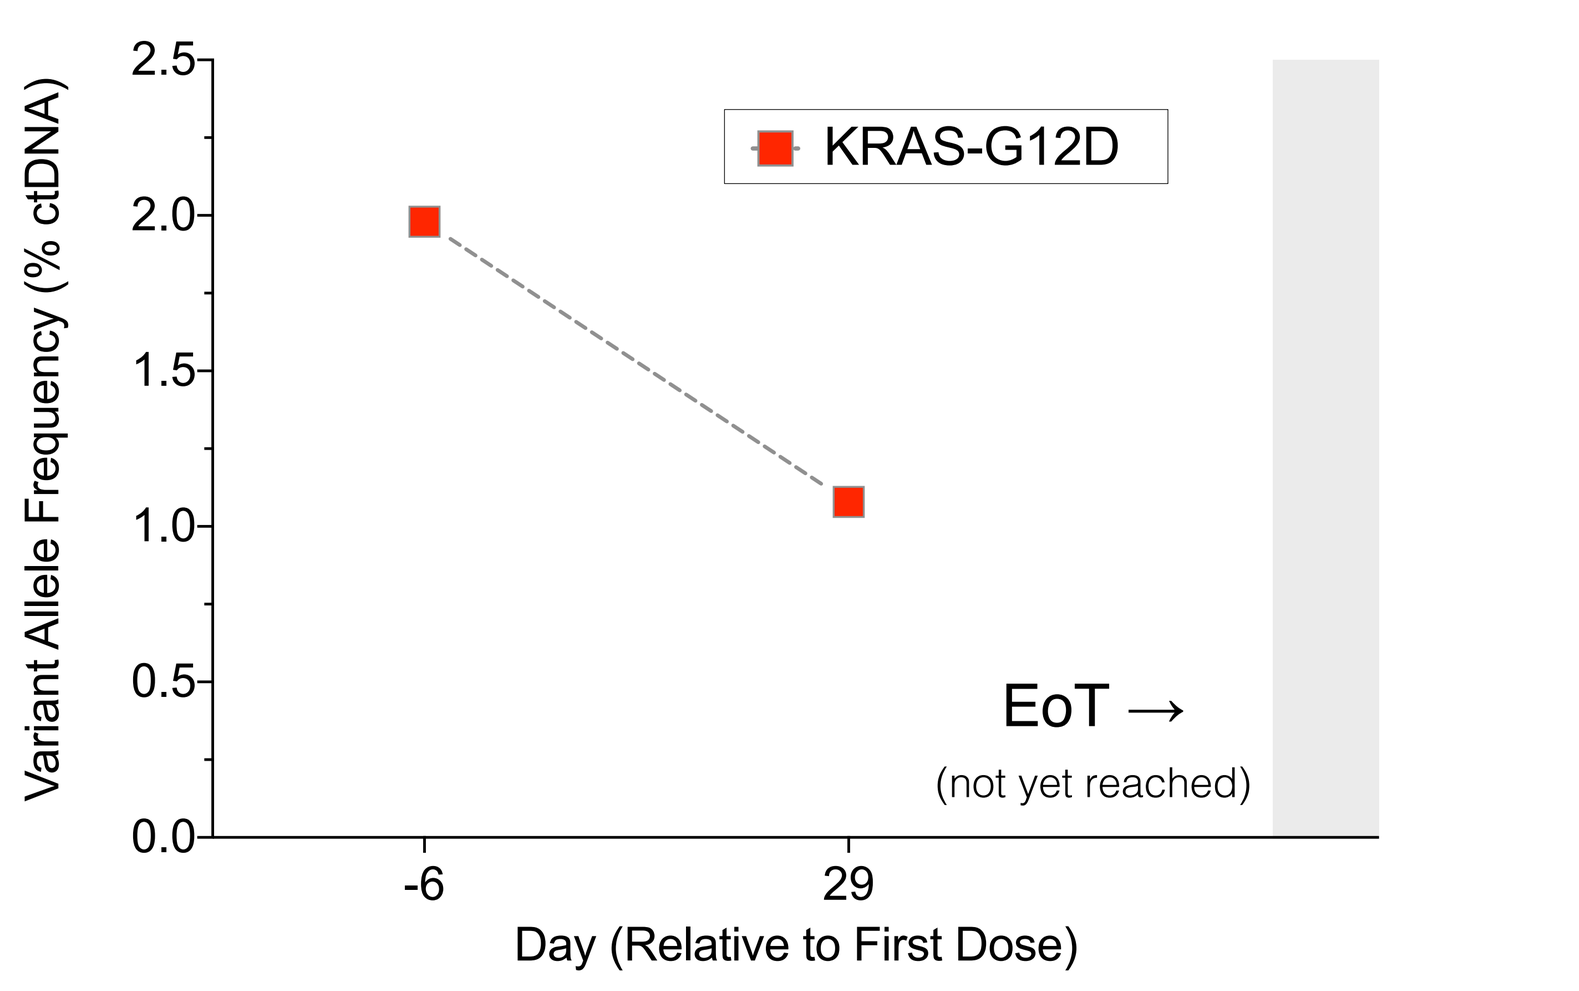
KRAS-G12D
Variant Allele Frequency (% ctDNA)
EoT →
(not yet reached)
Day (Relative to First Dose)
-6
29

Phase 1 Safety & Tolerability

IMM-1-104 safety data suggests that deep cyclic inhibition (DCI) of MEK is well tolerated at doses up to 240 and 320 mg QD p.o., where C_{max} drug free fractions typically exceeded 1,000 nM followed by daily, near-zero drug troughs. DCI was designed to maximally inhibit the MAPK pathway at MEK in a pulsatile fashion.

TOTAL TREATED PATIENTS (N=54)	Patients at 240 (N=19) or 320 mg (N=30) QD p.o.*				
Maximum Severity of TRAE's	**Grade 1**	**Grade 2**	**Grade 3 ** **	**Grade 4**	**Any Grade**
TRAE's occurring in ≥10% of patients n (%)					
Rash ***	8 (15)	5 (9)	1 (2)	0 (0)	14 (26)
Diarrhea	9 (17)	3 (6)	0 (0)	0 (0)	12 (22)
Nausea	8 (15)	1 (2)	0 (0)	0 (0)	9 (17)
Fatigue	6 (11)	3 (6)	0 (0)	0 (0)	9 (17)
Vomiting	6 (11)	2 (4)	0 (0)	0 (0)	8 (15)
Subretinal fluid	7 (13)	0 (0)	0 (0)	0 (0)	7 (13)
Retinopathy	3 (6)	3 (6)	0 (0)	0 (0)	6 (11)

* 54 treated patients also included 1 patient treated at 40 mg QD p.o., 1 patient treated at 80 mg QD p.o. and 3 patients treated at 160 mg QD p.o.
** All treatment-related adverse events (TRAE's), regardless of occurrence frequency or grade, were reversible and deemed non-serious
*** Preferred Terms Included in the Rash term include Dermatitis acneiform, Photosensitivity reaction, Rash, Rash macular, Rash maculo-papular, Rash pruritic, Rash pustular

Conclusions

- IMM-1-104 is a short-lived dual MEK inhibitor (MEKi) that is resistant to CRAF-bypass and has a unique pulsatile pharmacokinetic and pharmacodynamic mechanism (i.e., DCI MEKi)
- The primary objective of Phase 1 was to evaluate safety, tolerability and to identify a candidate recommended phase 2 dose (RP2D). A candidate RP2D was identified with a highly differentiated safety and tolerability profile.
- Pharmacodynamic inhibition of pERK upon testing human plasma samples *ex vivo* exceeded 90% at C_{max} for most patients dosed at 240 or 320 mg QD p.o., providing optimal IMM-1-104 DCI MEKi profiles[1].
- Antitumor activity was observed at both 240 and 320 mg QD p.o., as evidenced by deepening RECIST target lesion regressions as well as reductions in CA 19-9 and relevant ctDNA biomarkers.
- Emerging clinical data suggest that IMM-1-104 (i.e., DCI MEKi) provides class-differentiated activity and tolerability that warrants further evaluation in both monotherapy and select combinations[2].
- Drug efficacy will be further evaluated in Phase 2a. NCT05585320 is actively enrolling at 320 mg QD p.o. monotherapy in melanoma, lung and pancreatic cancer, and at 240 or 320 mg QD p.o. IMM-1-104 plus gemcitabine- or fluorouracil-containing chemotherapy in first-line pancreatic cancer patients.

References

1. Immuneering Corporation (2024, March 14). *Topline Results Phase 1 Portion.* Retrieved from https://ir.immuneering.com/news-releases/news-release-details/immuneering-announces-positive-topline-results-phase-1-portion
2. Quality of Life (QoL) assessed by overall improvement on Functional Assessment of Anorexia/Cachexia Treatment (FAACT) questionnaire (v4); DOI: 10.1023/a:1016670403148
3. P. King, et al. Activity of IMM-1-104 alone or in combination with chemotherapy in RAS-altered PDAC. 2024 Cancer Res 84 (6_Supplement): 4195

The authors would like to acknowledge all participating investigators, patients and caregivers. || Phase 1 database snapshot: August 19th, 2024.

[1]Presenting author: Vincent Chung, M.D. [VChung@coh.org] has no conflicts of interest. Study sponsored by Immuneering Corporation (employees are stockholders of Immuneering Corporation). Sponsor contact: Brett Hall - bhall@Immuneering.com
Presented ESMO Annual Conference 2024, Barcelona, Spain (September 16th, 2024) | FPN: 1524P
Immuneering